E17

Evaluation of Submicron Particle Docetaxel Directly Injected into Uro-Oncologic Xenografts

Gere diZerega[1,2,3], Holly Maulhardt[2], Sara Dafoe[2], Lauren Peterson[2], Michael Frost[4], Ashley Tornio[2], Leanne Drummond[2]
[1]Keck School of Medicine, USC, Los Angeles, CA; [2]US Biotest, Inc., San Luis Obispo, CA; [3]NanOlogy, LLC, Fort Worth, TX; [4]Western Diagnostic Services Laboratory, Santa Maria, CA

Genitourinary Cancers Symposium
Translating Evidence to Multidisciplinary Care

ABSTRACT #360

BACKGROUND: Suspensions of submicron particle taxanes (Figure 1) allow for direct administration of high, sustained levels of chemotherapeutics at the site of disease. Intratumoral (IT) injections of submicron particles of pure docetaxel (NanoDoce) into clear cell renal carcinoma (786-O), transitional cell bladder carcinoma (UM-UC-3) and prostate carcinoma (PC-3) xenografts were evaluated.

METHODS: Tumor cells (786-O and UM-UC-3) or tumor fragments (PC-3) were implanted subcutaneously into the hind flank of immunocompromised rats or mice. Treatments were initiated at 7 days (786-O, Figure 3), 18 days (UM-UC-3, Figure 5) and 26 days (PC-3, Figure 7) following implant. Vehicle, IT NanoDoce, and intravenous (IV) docetaxel treatments were administered on weekly cycles per Table 1. Animals were followed up to 60 days post-treatment initiation. Tumor size was measured using calipers and body weights were collected 2-3 times per week. In the 786-O and UM-UC-3 studies, tumor site tissues were analyzed for docetaxel levels using LC-MS/MS (Figure 2) and hematoxylin & eosin (H&E) and immunohistochemistry (IHC) (Pan-Cytokeratin AE-1/AE-3, CD11b, CD16, and CD68) was evaluated (Figures 4 and 6).

RESULTS: Tumor volume reductions with 2 and 3 doses of NanoDoce were significantly greater than (renal and bladder) or similar (prostate) to IV docetaxel and significantly greater than vehicle ($p<0.05$). Histology confirmed tumor volume findings; significant tumor regression with increased immune-cell infiltrate observed in IT NanoDoce treatments compared to controls. High tissue levels of docetaxel were detected in NanoDoce-treated animals. IHC showed (peri)tumor-infiltrating immune cells in NanoDoce-treated animals while vehicle and IV docetaxel had limited to no infiltration.

Figure 1. Submicron Particle Production Technology

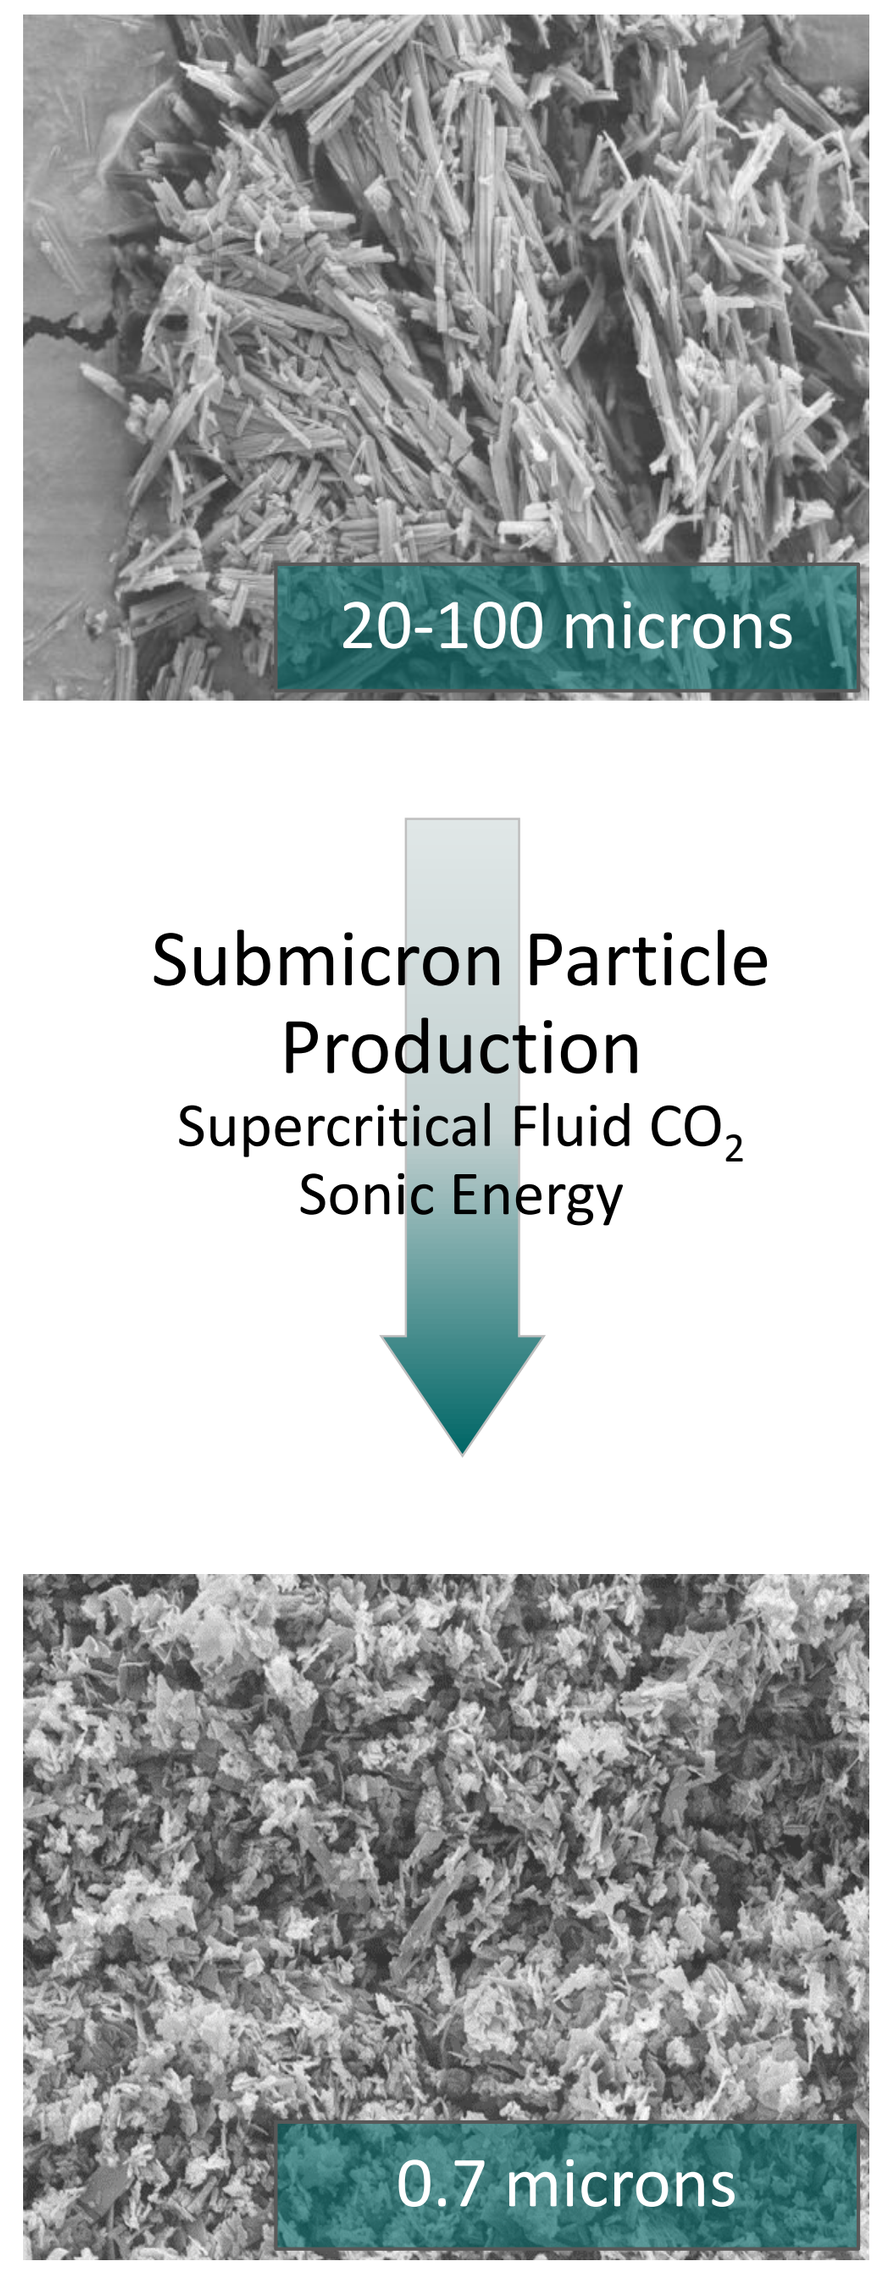
20-100 microns
Submicron Particle Production
Supercritical Fluid CO_2
Sonic Energy
0.7 microns

CritiTech, Inc.; Lawrence, KS

- Large active pharmaceutical ingredient (API) crystals cannot be suspended and must be dissolved for IV delivery
- Small amount of systemically delivered dose reaches the tumor site for a short period of time
- Each submicron particle contains 2-3 billion molecules of taxane
- Particles suspended and locally delivered into tumor are entrapped at disease site for long durations
- Increased particle surface area allows for sustained therapeutic API release

NanoDoce® is a registered trademark of NanOlogy, LLC.

Table 1. Study Designs

Indication/ Cell Line	Xenograft Model (n)	Treatment	Dose (mg/kg)	# Weekly Cycles
Renal/ 786-O	Sprague-Dawley *OncoRat® SRG™* (*Rag2/Il2rg* null) (30)	IT vehicle	N/A	3
		IV docetaxel	5.0 - 2.5	2 or 3
		IT NanoDoce	20	1, 2, or 3
Bladder/ UM-UC-3	Hsd:Athymic Nude-*Foxn1^{nu}* mice (47)	IT vehicle	N/A	3
		IV docetaxel	30	3
		IT NanoDoce	100	1, 2, or 3
Prostate/ PC-3	Crl:NU(NCr)-*Foxn1^{nu}* mice (50)	IT vehicle	N/A	3
		IV docetaxel	30	3
		IT NanoDoce	37.5	3
			100	1 or 3

Figure 2. Docetaxel Tumor Residence Time

- Compared to IV docetaxel, concentrations (µg docetaxel/gram tumor) were much greater in renal and bladder tumors following IT NanoDoce-treatment
- Docetaxel in tumor tissues was detected up to 50 days after IT administration of NanoDoce

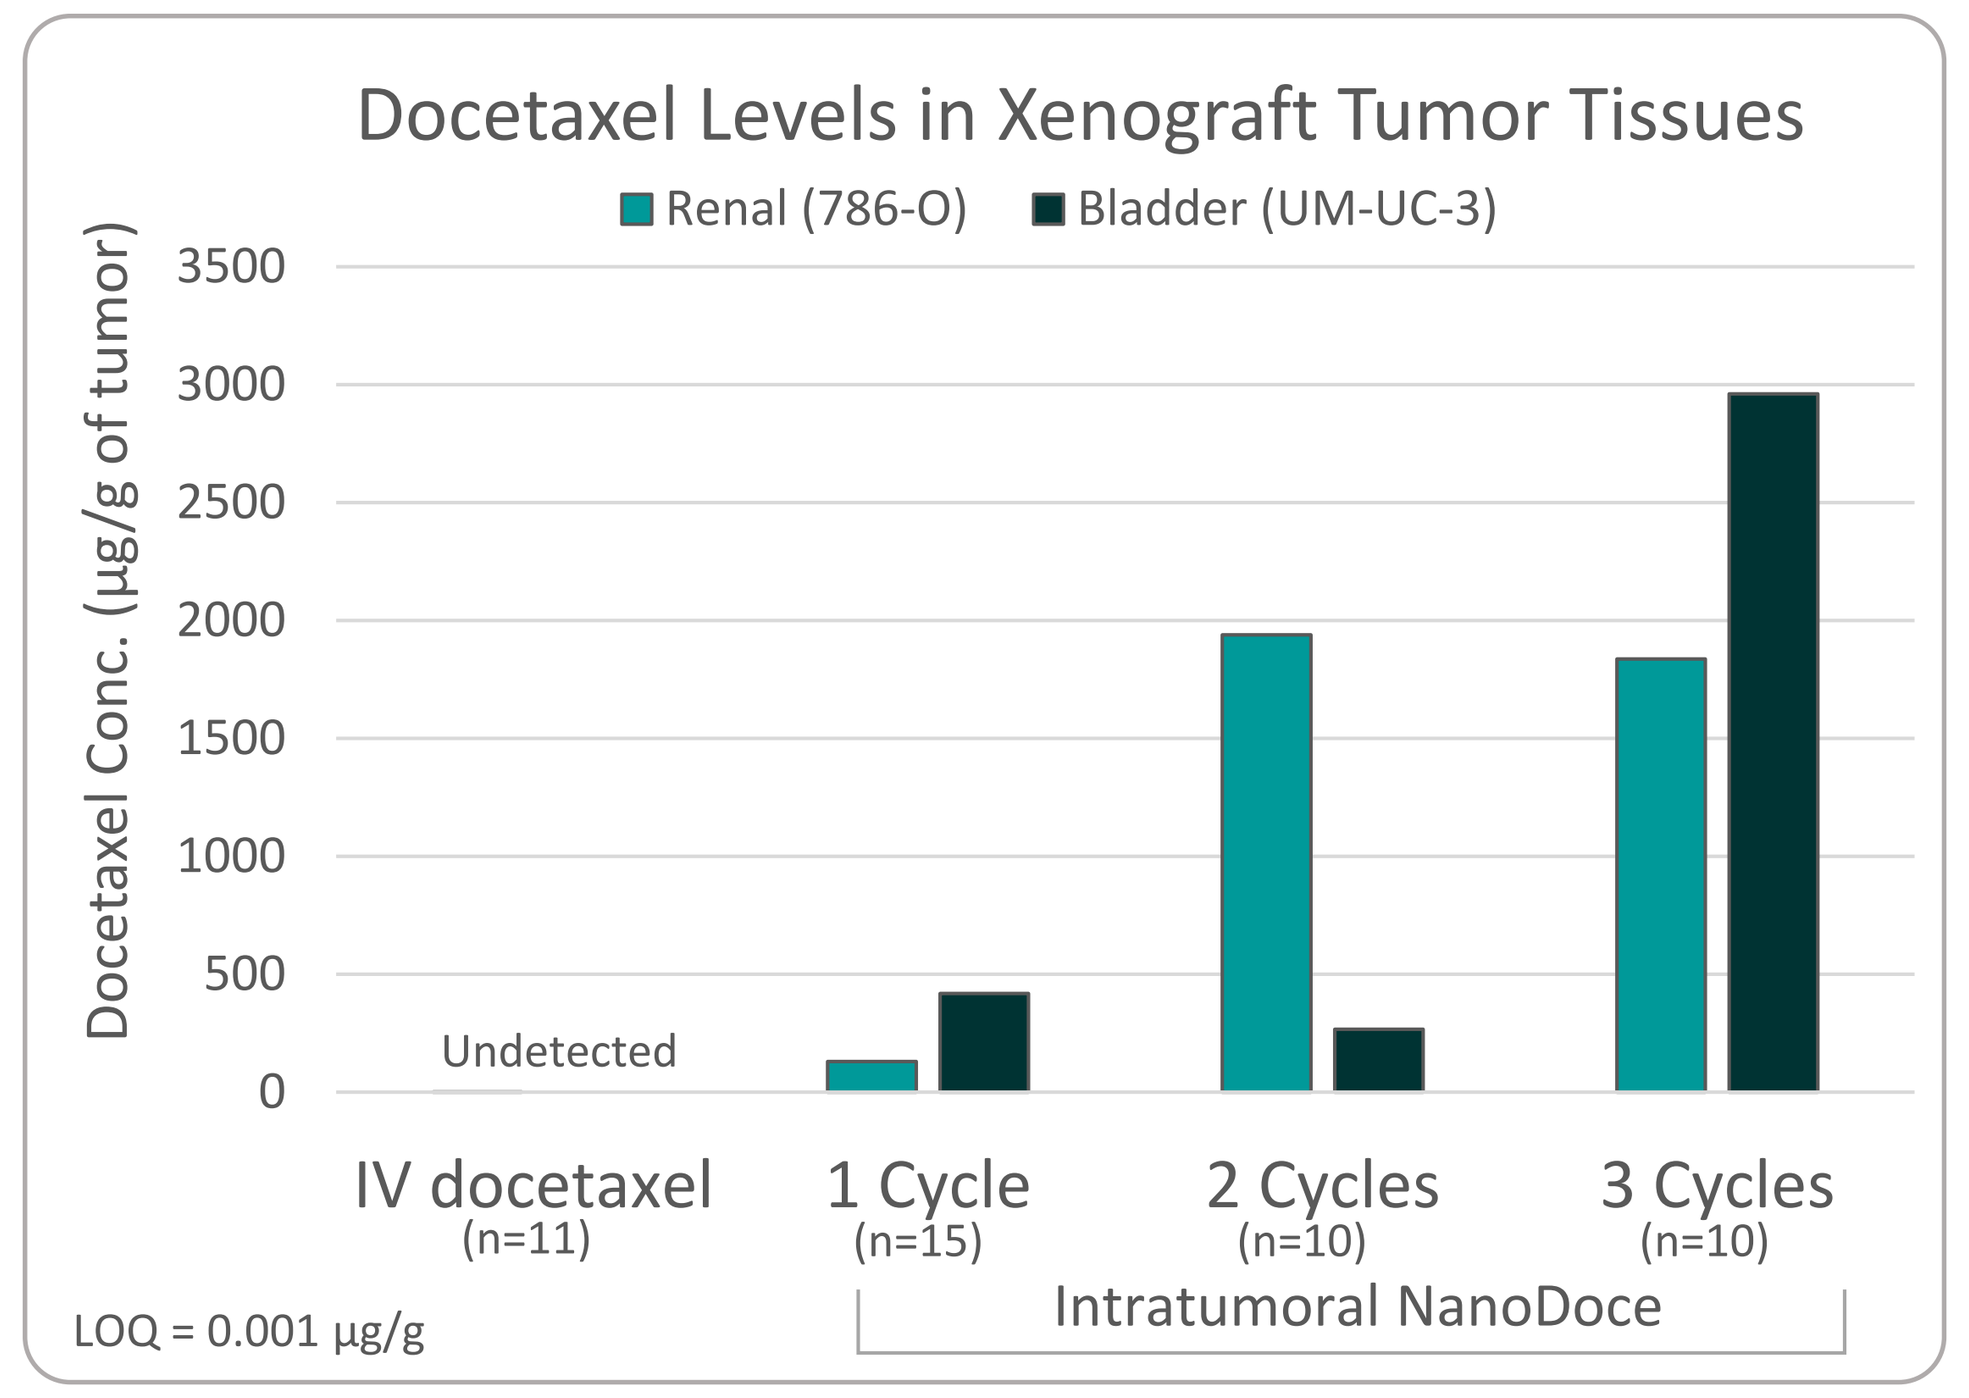
Docetaxel Levels in Xenograft Tumor Tissues
Renal (786-O)
Bladder (UM-UC-3)
Docetaxel Conc. (µg/g of tumor)
3500
3000
2500
2000
1500
1000
500
0
Undetected
IV docetaxel (n=11)
1 Cycle (n=15)
2 Cycles (n=10)
3 Cycles (n=10)
Intratumoral NanoDoce
LOQ = 0.001 µg/g

Tumors were collected 25 to 36 days (IV docetaxel), 28 to 50 days (1 Cycle), 37 to 43 days (2 Cycles), or 30 to 36 days (3 Cycles) after final treatment.

Figure 3. Clear Cell Renal Carcinoma Xenograft

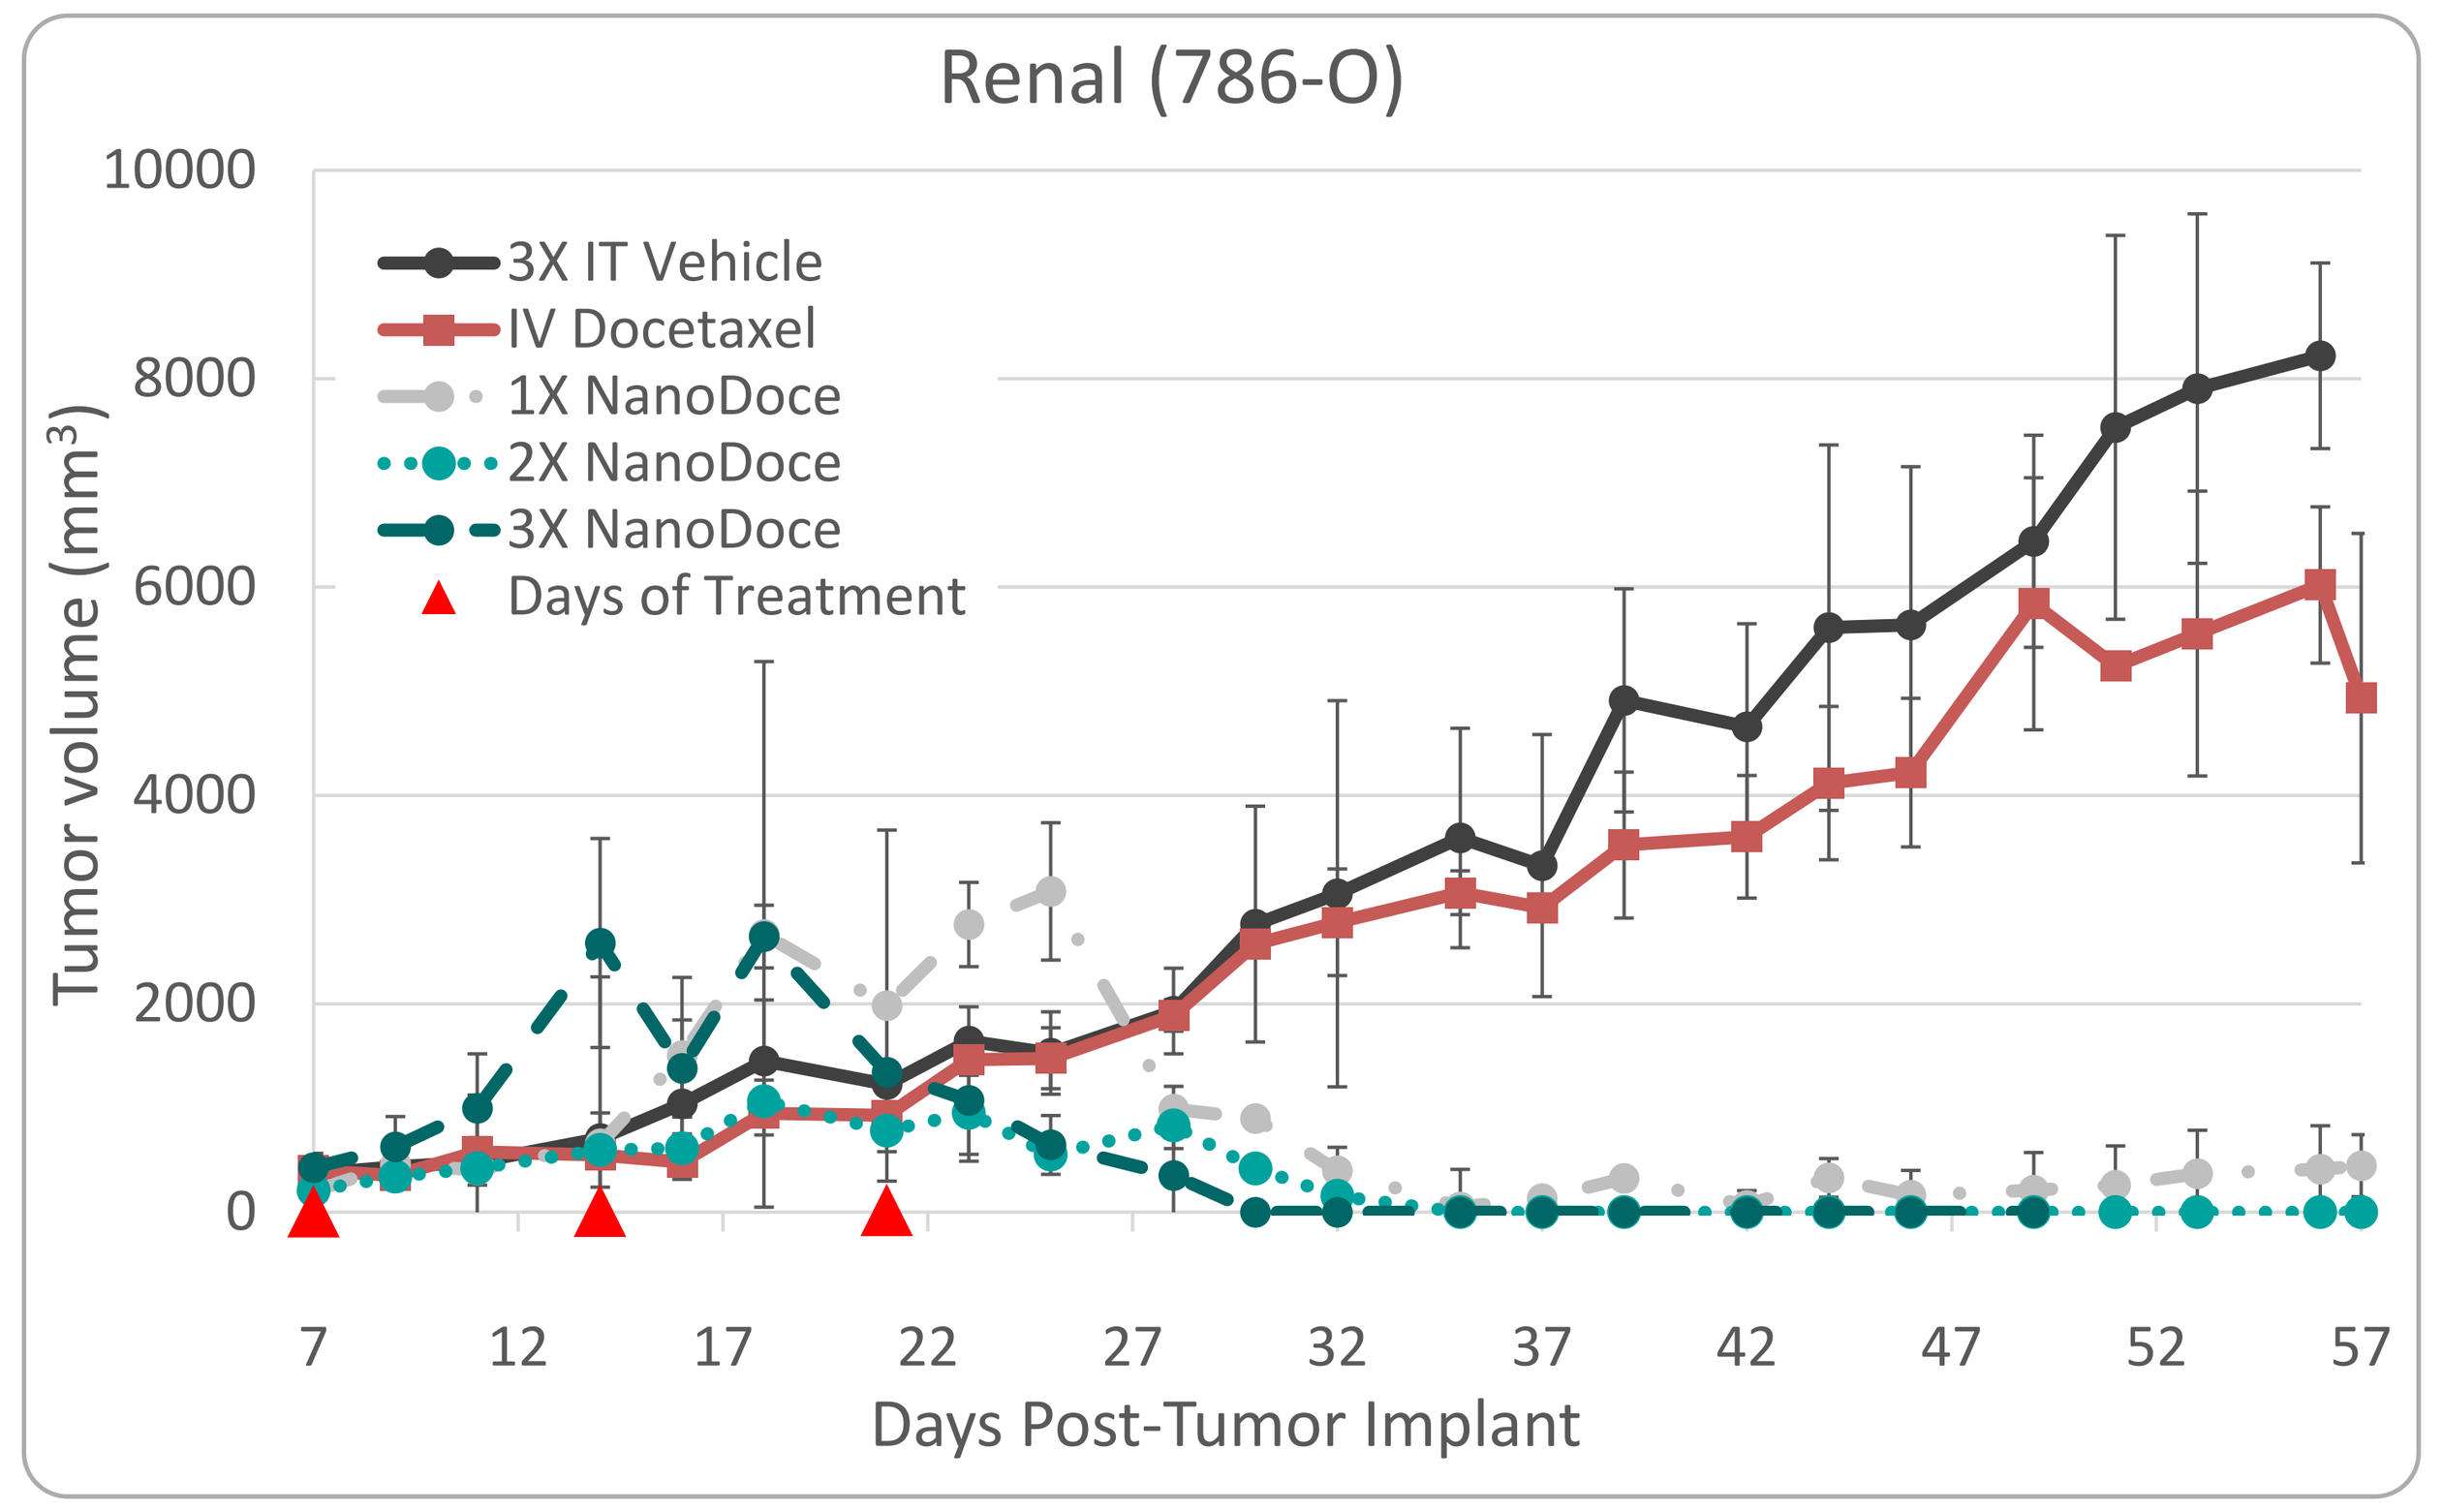
Renal (786-O)
Tumor volume (mm³)
10000
8000
6000
4000
2000
0
3X IT Vehicle
IV Docetaxel
1X NanoDoce
2X NanoDoce
3X NanoDoce
Days of Treatment
7
12
17
22
27
32
37
42
47
52
57
Days Post-Tumor Implant

Figure 4. Clear Cell Renal Carcinoma Histology

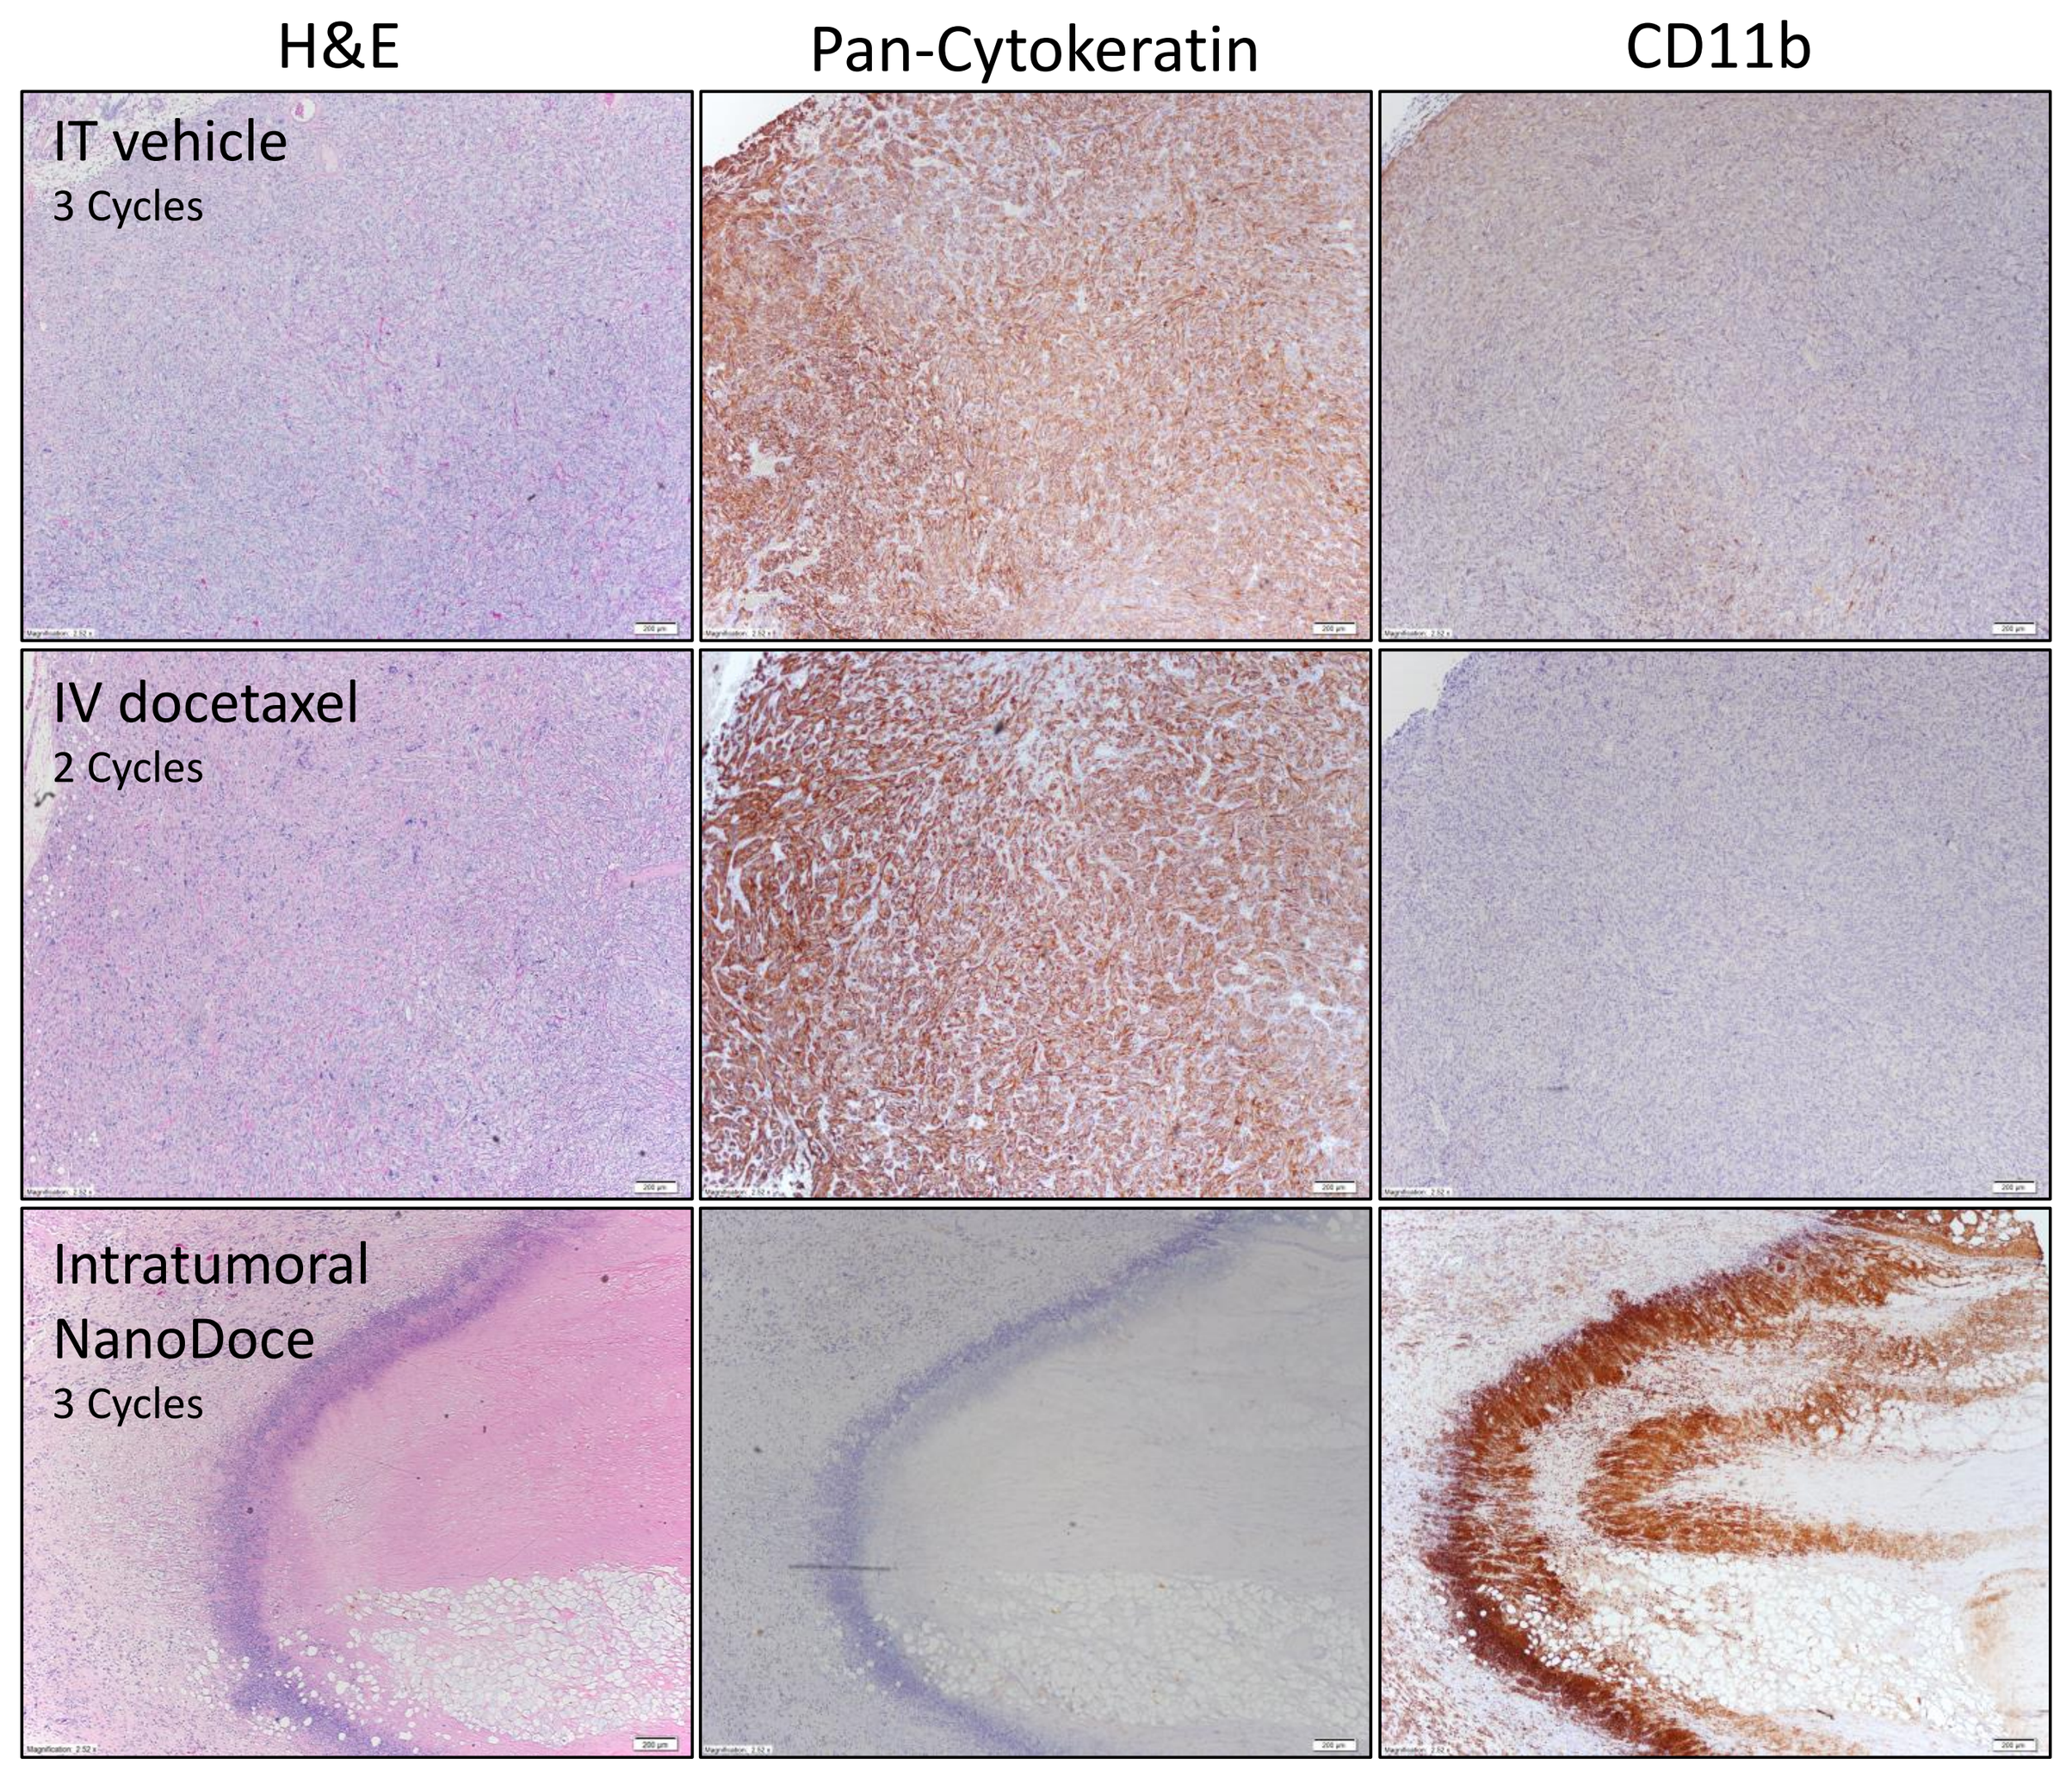
H&E
Pan-Cytokeratin
CD11b
IT vehicle
3 Cycles
IV docetaxel
2 Cycles
Intratumoral
NanoDoce
3 Cycles

Paraffin-embedded tumor site tissues were stained with H&E and IHC was performed (pan-cytokeratin and CD11b). Vehicle and IV docetaxel tissues were collected 36 days and intratumoral NanoDoce tissues were collected 28 days after final treatments. Row 1 vehicle and Row 2 IV docetaxel: tumors contain dense invasive carcinoma composed of closely packed viable tumor cells confirmed with cytokeratin stain. CD11b+ staining is sparse and minimal. Row 3 intratumoral NanoDoce: tissues show extensive tumor cell coagulative necrosis with no residual viable invasive carcinoma noted in H&E and pan-cytokeratin. NanoDoce treated tumor sites have marked CD11b+ infiltrate stained within the necrotic tissue. Magnification: 2.52x

Renal Cell Carcinoma Clinical Trial

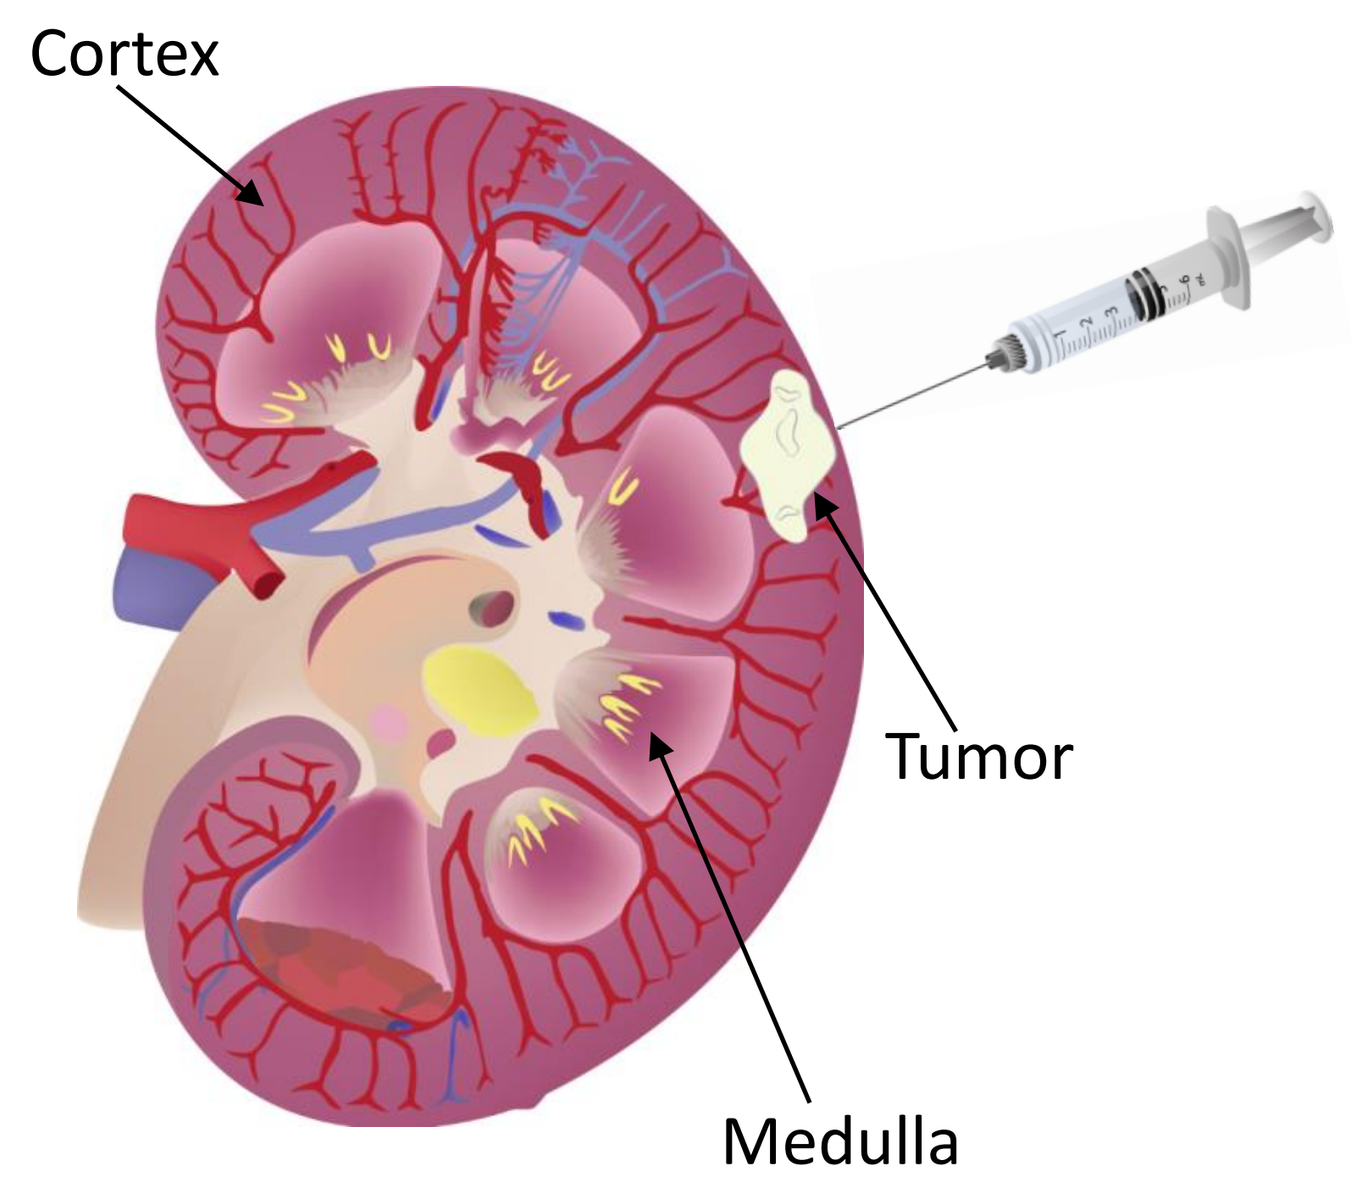
Cortex
Tumor
Medulla

- Ultrasound-guided direct injection of NanoDoce into 1.5 – 4.0 cm localized renal tumors
- GLP Toxicology Study in-process

Figure 5. Transitional Cell Bladder Carcinoma Xenograft

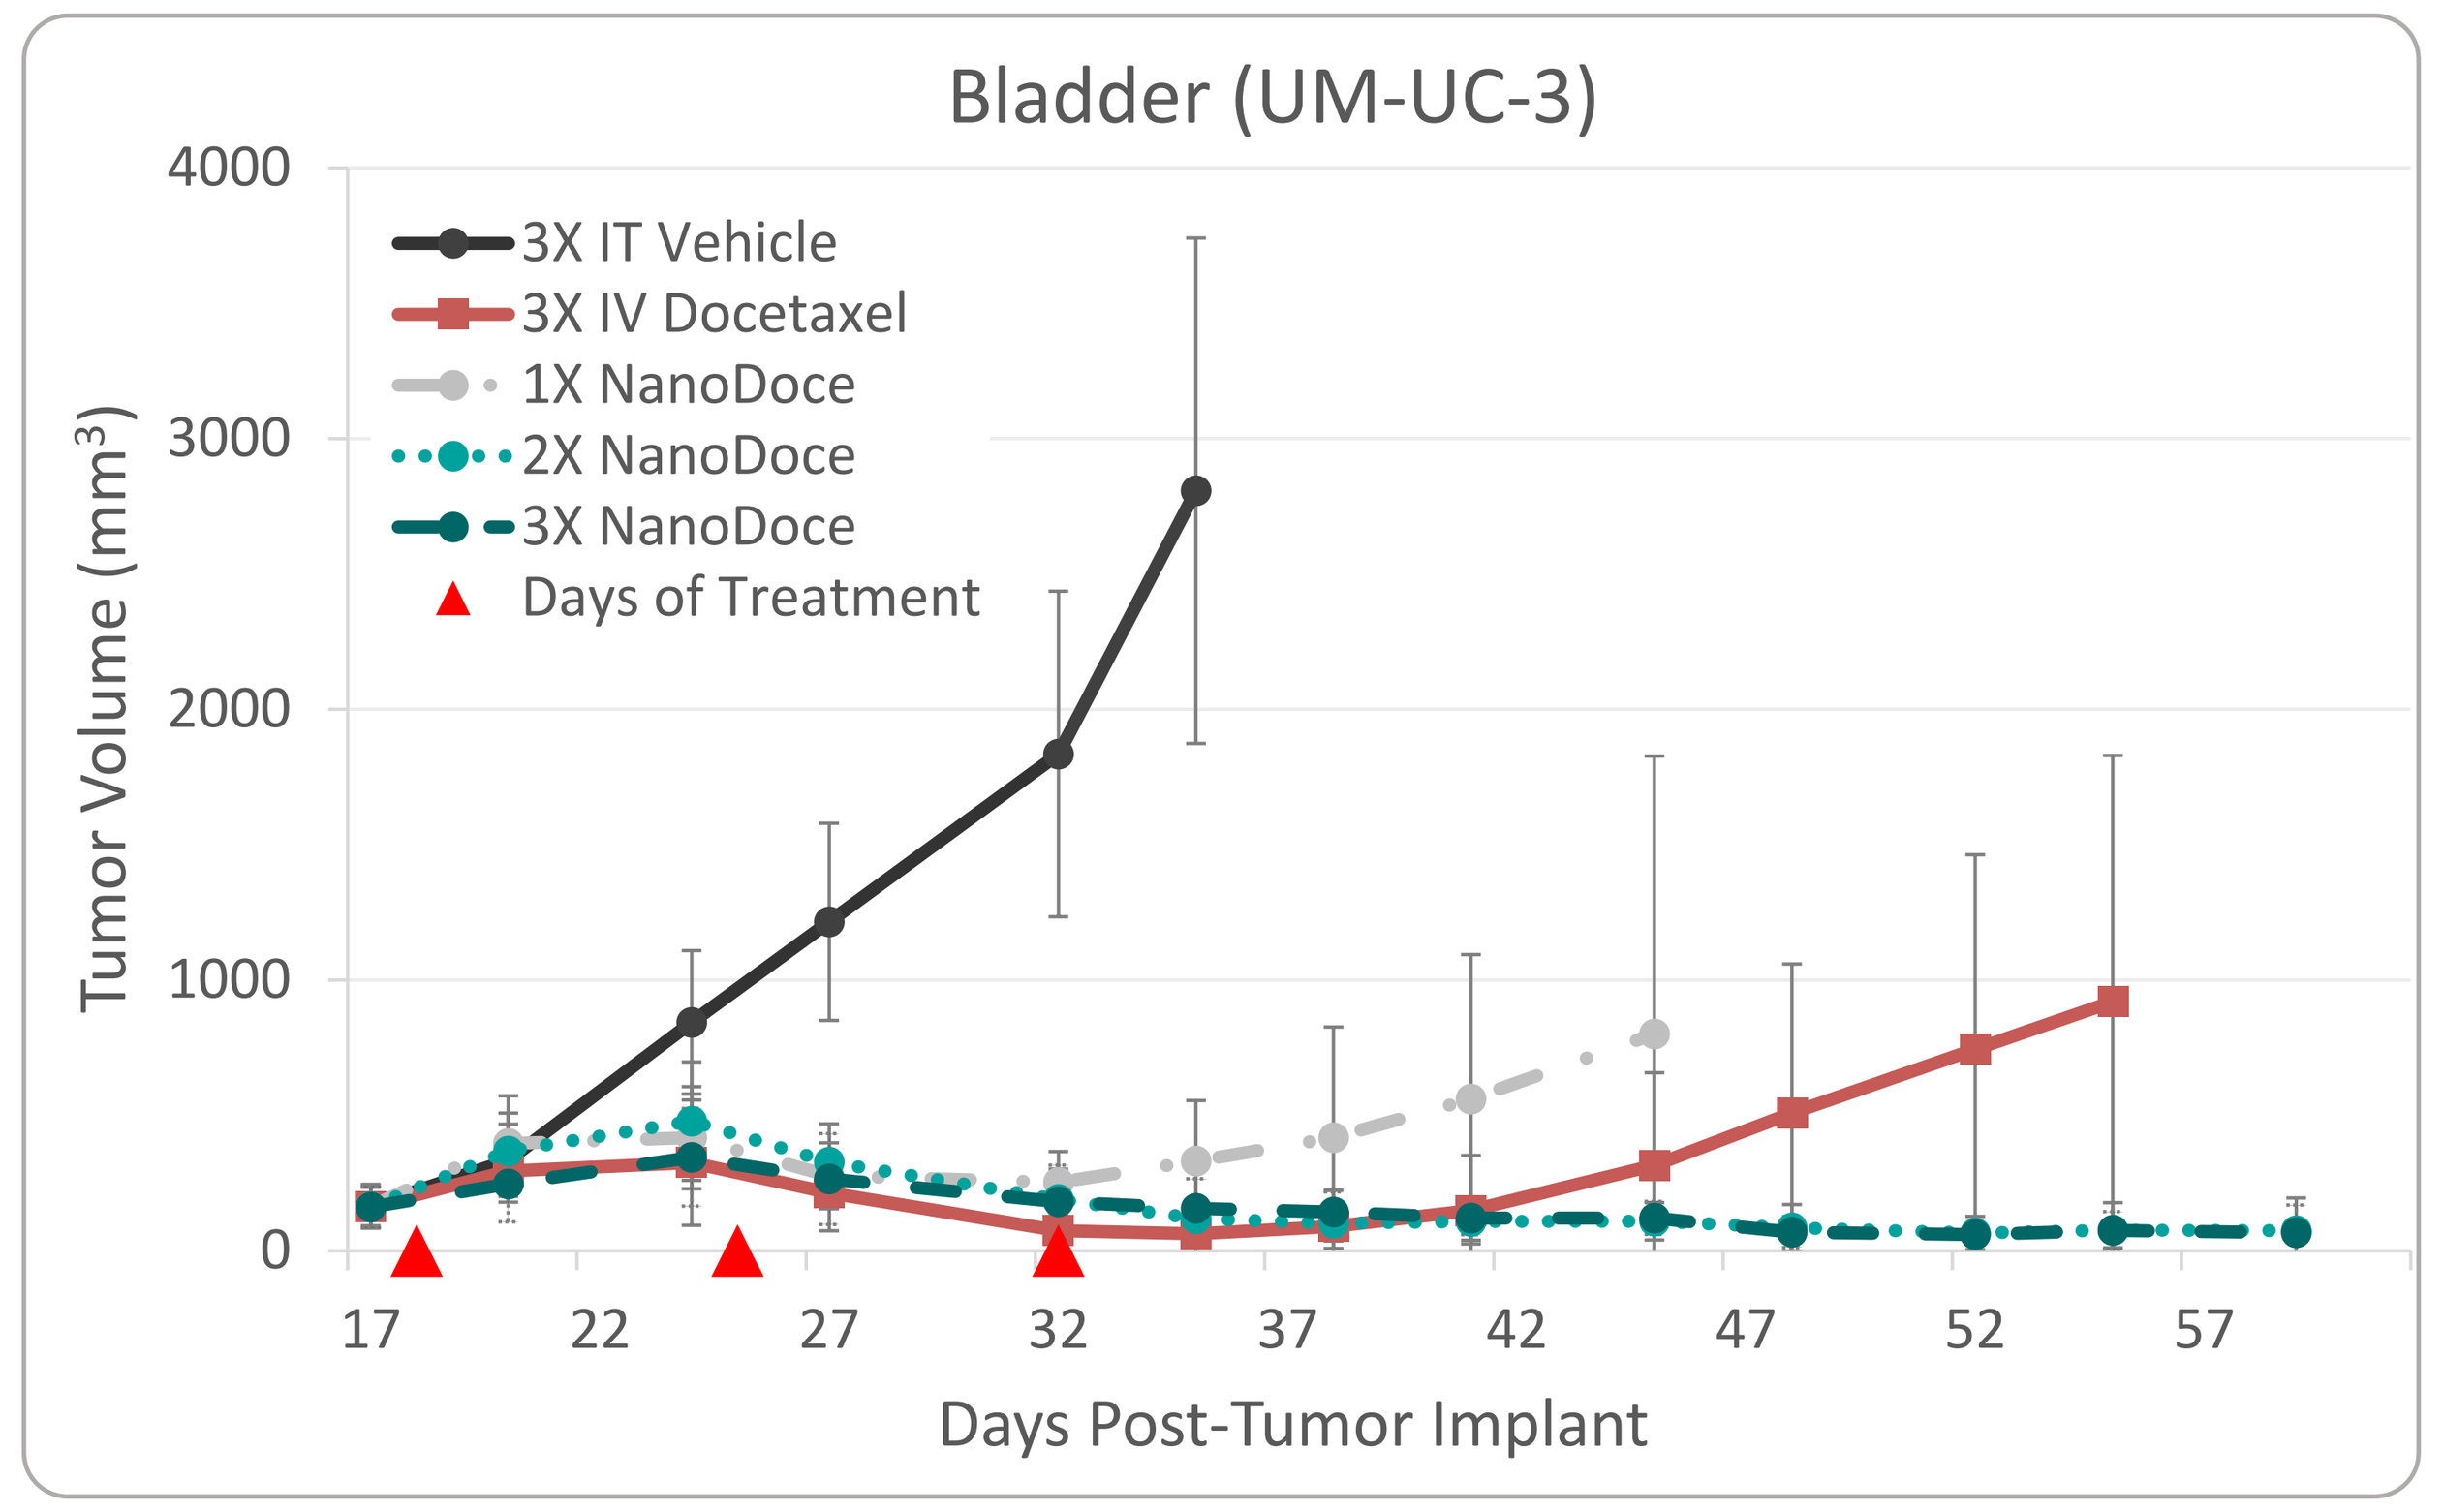
Bladder (UM-UC-3)
Tumor Volume (mm³)
4000
3000
2000
1000
0
3X IT Vehicle
3X IV Docetaxel
1X NanoDoce
2X NanoDoce
3X NanoDoce
Days of Treatment
17
22
27
32
37
42
47
52
57
Days Post-Tumor Implant

Figure 6. Transitional Cell Bladder Carcinoma Histology

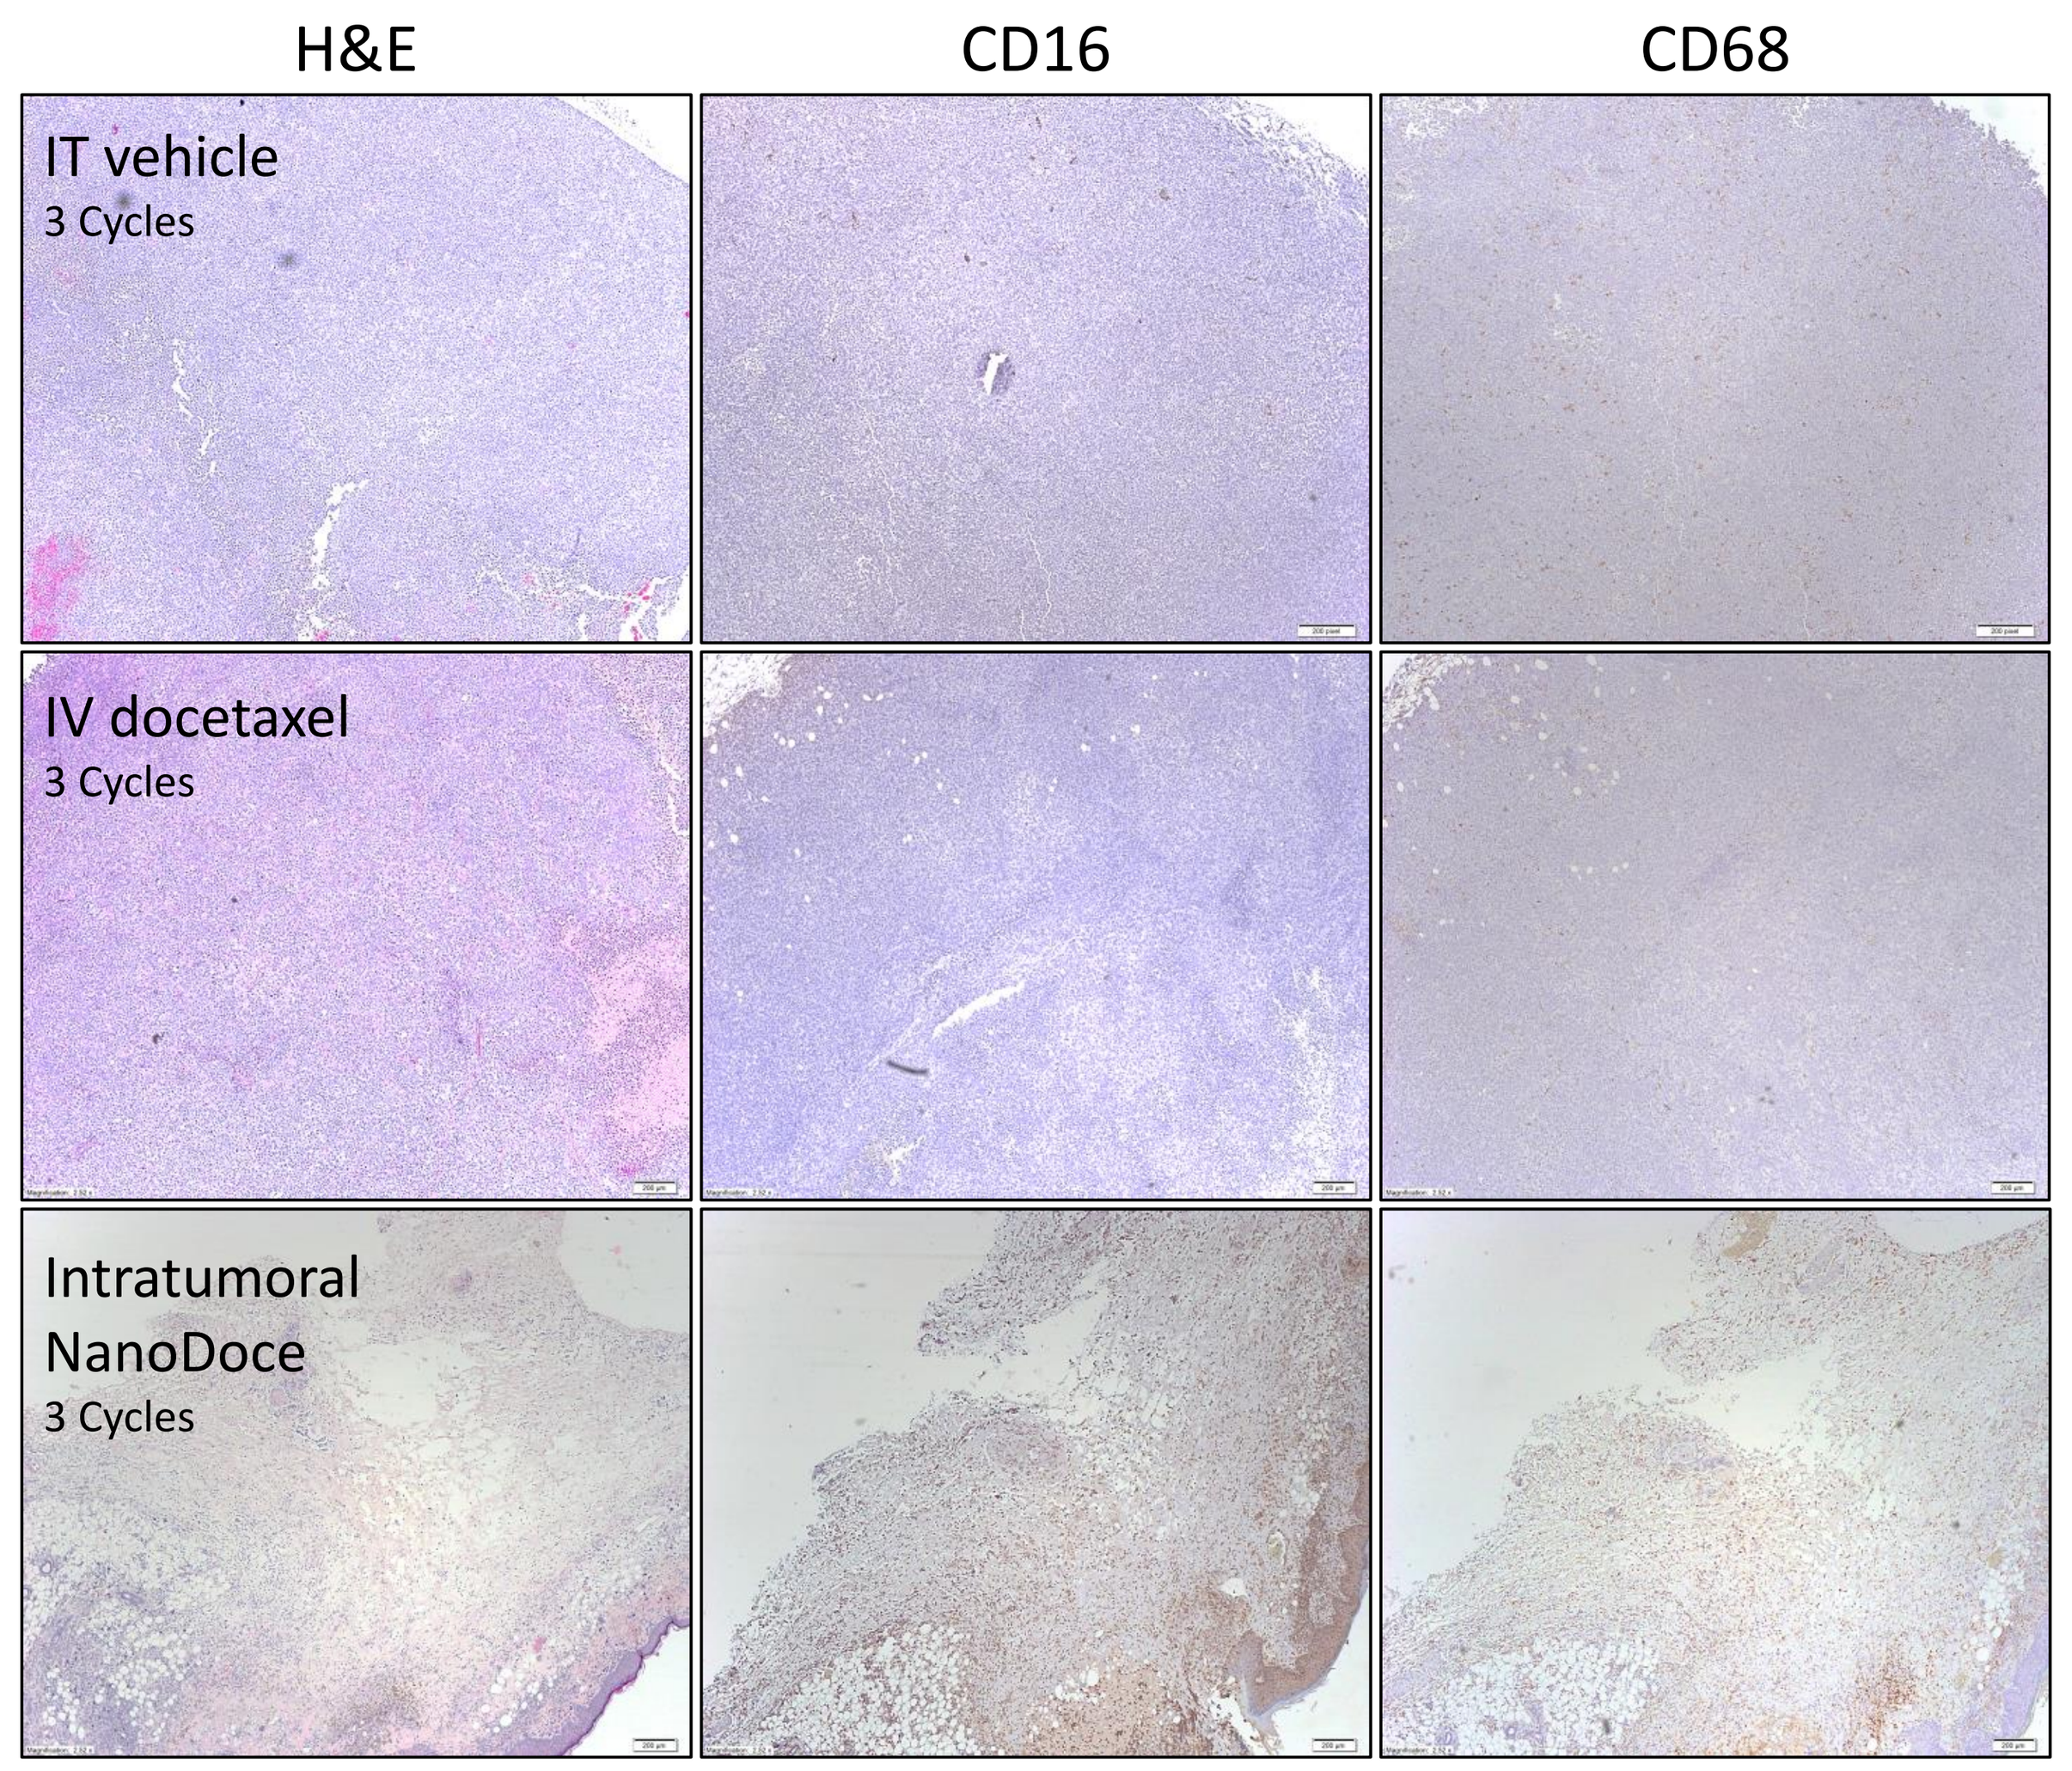
H&E
CD16
CD68
IT vehicle
3 Cycles
IV docetaxel
3 Cycles
Intratumoral
NanoDoce
3 Cycles

Paraffin-embedded tumor site tissues were stained with H&E and IHC was performed (CD16 and CD68). Vehicle, IV docetaxel, and intratumoral NanoDoce tissues were collected 30 days after final treatments. Row 1 vehicle and Row 2 IV docetaxel: extensive viable invasive carcinoma composed of sheets of closely-packed tumor cells. No CD16+ (NKs) infiltrate present and mild CD68+ macrophage infiltrate confirmed. No significant lymphoid infiltrate within vehicle or IV docetaxel treated groups. Row 3 intratumoral NanoDoce: extensive tumor cell necrosis observed with no residual viable carcinoma present, moderate CD16+ and moderate stromal CD68+ stained immune-cell infiltrate present. Magnification: 2.52x

Bladder Cancer Clinical Trial

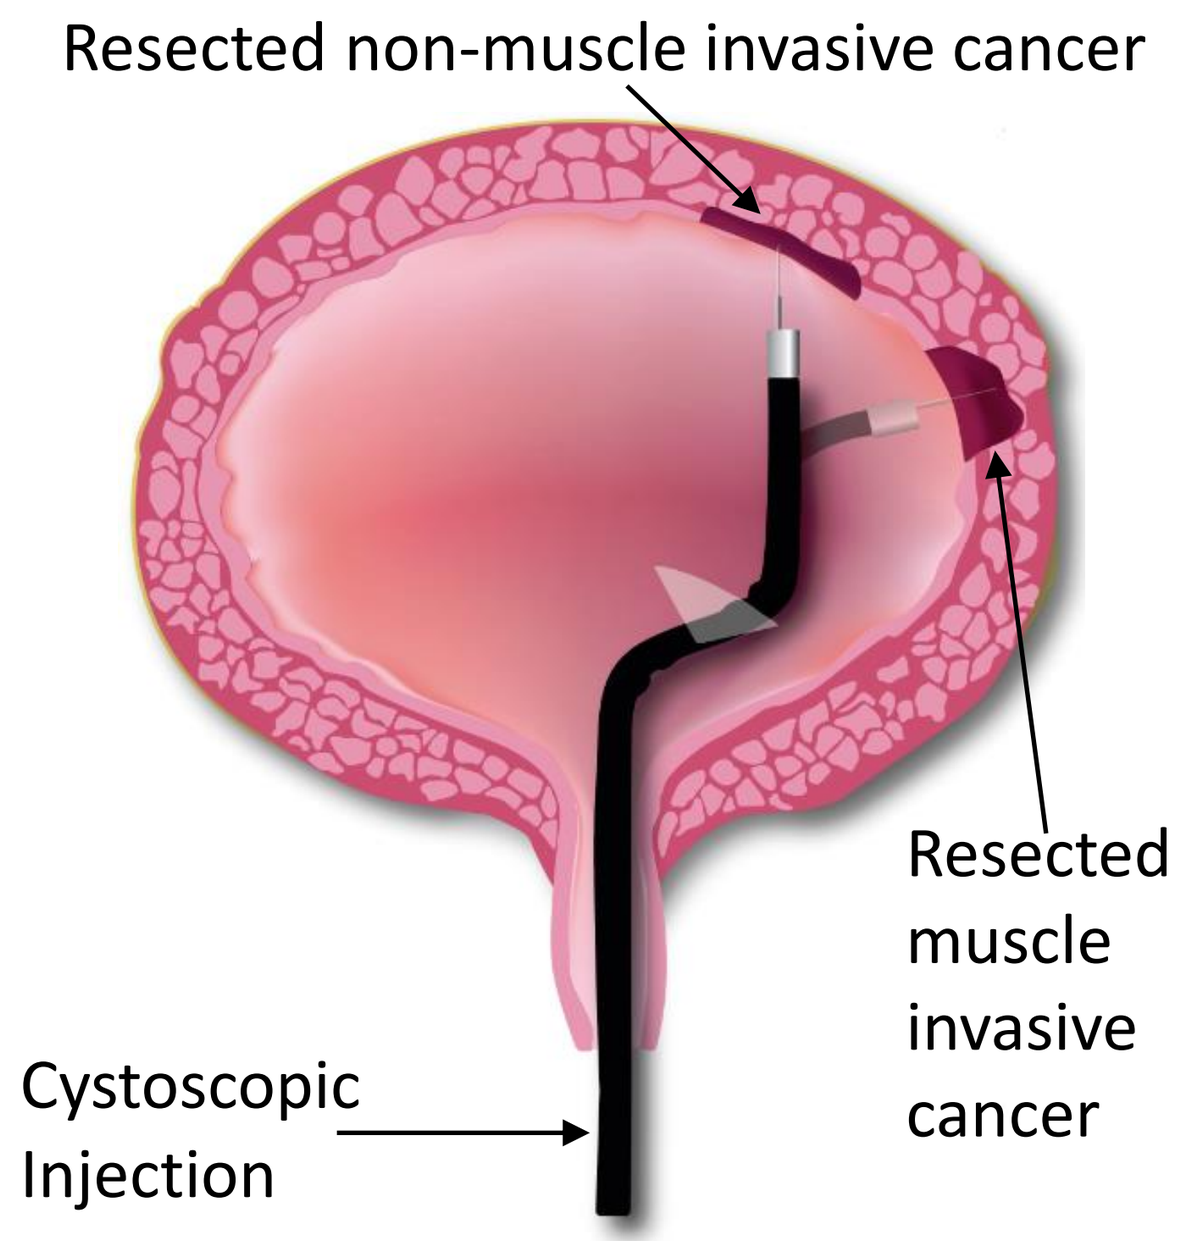
Resected non-muscle invasive cancer
Resected muscle invasive cancer
Cystoscopic Injection

- Under image-guidance, subjects with non-muscle invasive bladder cancer (NMIBC; BCG-unresponsive disease and/or high-risk) or muscle invasive bladder cancer (MIBC) receive NanoDoce as direct injection to the tumor resection site and intravesical instillation
- GLP Toxicology Studies completed
- Initiation of trial sites ongoing
- Clinical Trial #NCT03636256

Figure 7. Prostate Carcinoma Xenograft

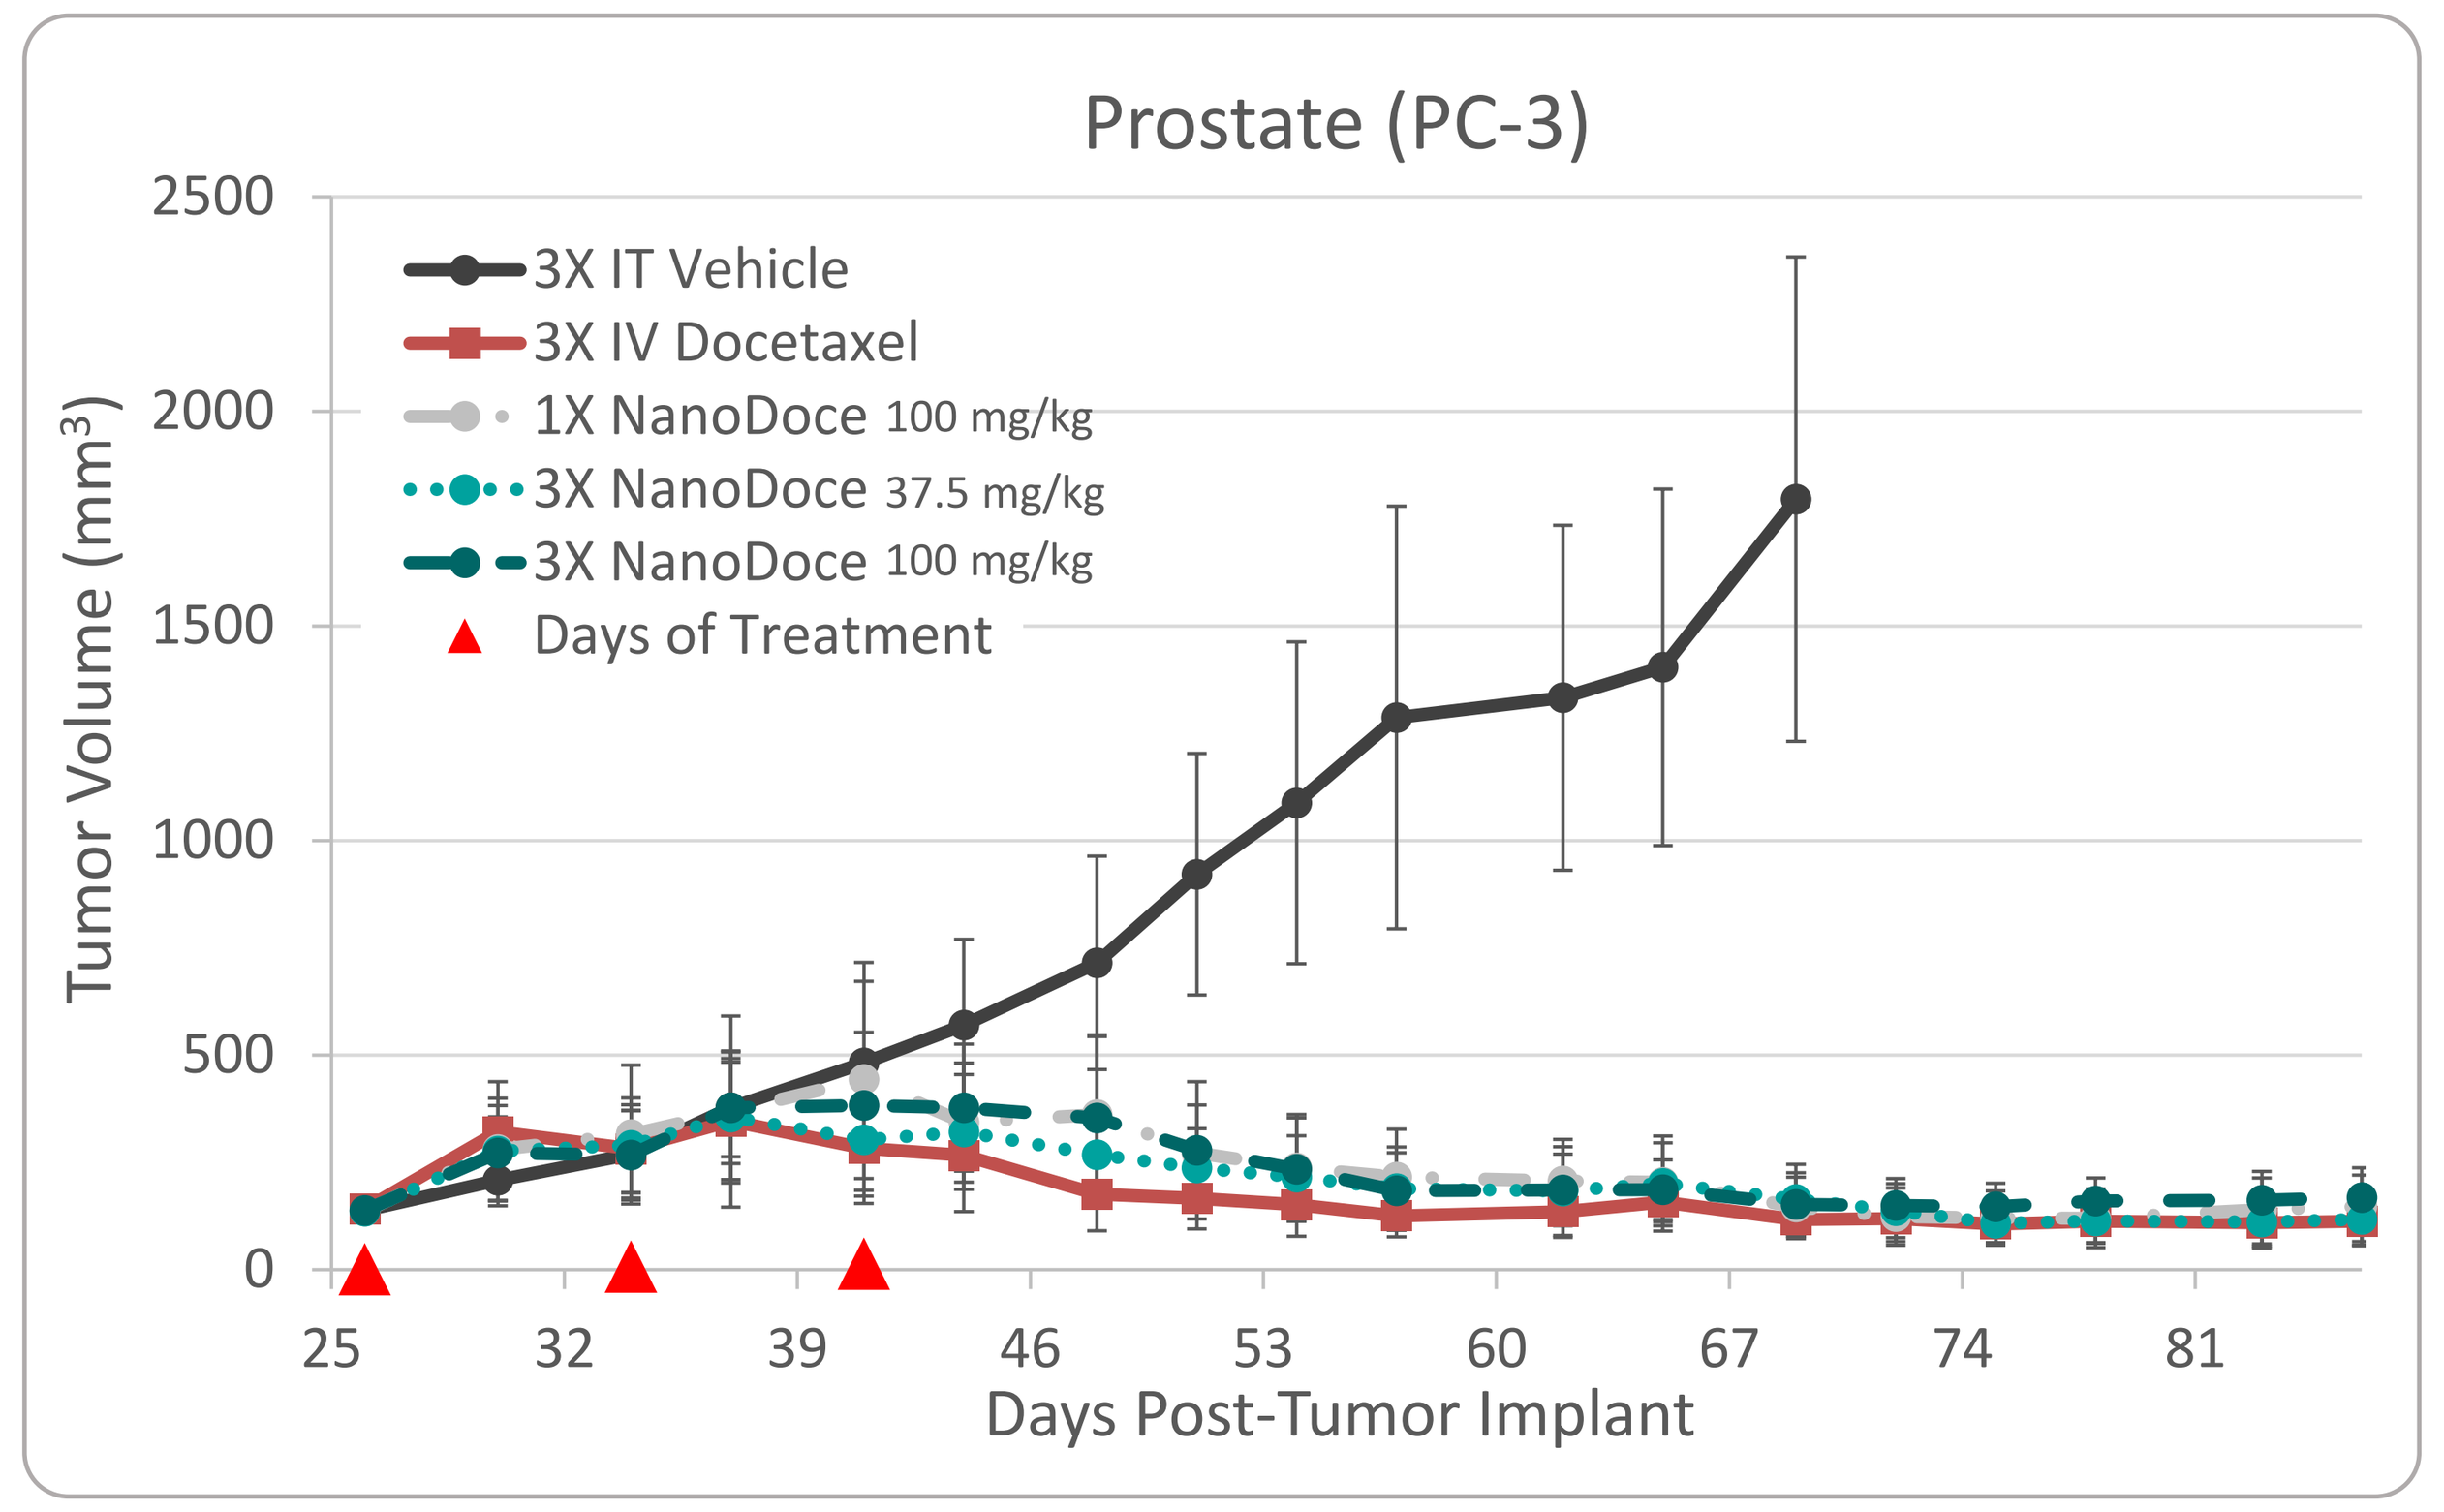
Prostate (PC-3)
Tumor Volume (mm³)
2500
2000
1500
1000
500
0
3X IT Vehicle
3X IV Docetaxel
1X NanoDoce 100 mg/kg
3X NanoDoce 37.5 mg/kg
3X NanoDoce 100 mg/kg
Days of Treatment
25
32
39
46
53
60
67
74
81
Days Post-Tumor Implant

Summary and Conclusions

- Local injections of submicron particle docetaxel (NanoDoce) resulted in tumor reduction in three different models of uro-oncologic cancers and produced a unique disruption of the tumor microenvironment
- Treatment with multiple cycles of IV docetaxel resulted in minimal renal tumor volume reduction or unsustainable bladder tumor volume reduction
- Intratumoral NanoDoce administration provided high docetaxel levels within the tumor; orders of magnitude greater than IV administration
- Histologically, intratumoral injections of NanoDoce demonstrated high levels of tumor regression and increased local immune cell infiltrates compared to IV docetaxel treatment
- IT injection of NanoDoce may kill tumor cells through direct and indirect means
 - NanoDoce directly inhibits tumor cell mitosis
 - In reducing tumor burden, NanoDoce indirectly promotes immune cell-mediated tumor clearance
- Clinical testing of submicron particle taxanes in uro-oncologic cancers is underway

Prostate Clinical Trial

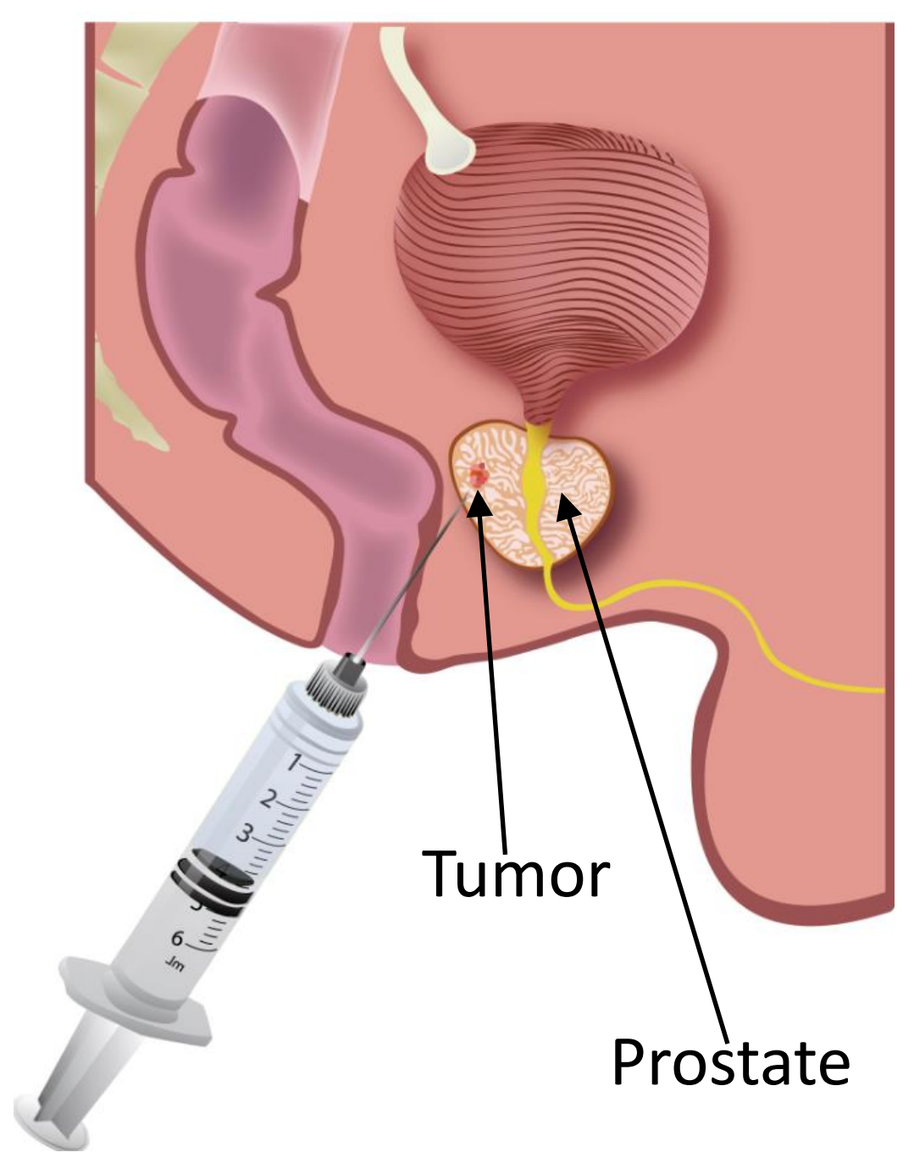
Tumor
Prostate

- Under image-guidance, submicron particle paclitaxel was injected directly into the lobe of the prostate with the dominant lesion 4 weeks prior to prostatectomy
- GLP Toxicology Study completed
- Clinical Trial #NCT03077659 Completed

Contact
Gere diZerega, MD gere.dizerega@usbiotest.com
NanOlogy.us