P82

Enhanced Tumor Regression and Immune Cell Infiltration by Inhaled Submicron Particle Paclitaxel in an Orthotopic Athymic Nude Rat Model of Non-Small Cell Lung Cancer

William Johnston[1], James Verco[1], Gere diZerega[1,2], Michael Frost[5], Michael Baltezor[2,3], Philip Kuehl[4]

[1]US Biotest, Inc., San Luis Obispo, CA, USA; [2]Nanology, LLC., Fort Worth, TX, USA; [3]CritiTech, Inc., Lawrence, KS, USA; [4]Lovelace Biomedical, Albuquerque, NM, USA; [5]Western Diagnostic Services Laboratory, Santa Maria, CA, USA

ATS 2019 Where today's science meets tomorrow's care™

Abstract (#8156)

Background: NanoPac consists of pure (uncoated) submicron particles of paclitaxel (Figure 1). In a previous study, healthy male rats received a single exposure of inhaled nebulized NanoPac or intravenous (IV) nab-paclitaxel. $T_{1/2}$ of inhaled NanoPac or IV nab-paclitaxel were 56 hrs or 20 hrs, with paclitaxel concentrations in the lung quantifiable 14- or 3-days post-administration, respectively (Figures 2 and 3)[1]. Lung tissue sampled at final necropsy was microscopically indistinguishable from untreated controls. The study data described here confirms and extend the therapeutic effects inhaled NanoPac previously reported[2,3].

Materials and Methods: 120 x-irradiated nude rats (n=20/group; Table 1) were intratracheally instilled with 20 x 10^6 Calu-3 non-small cell lung cancer (NSCLC) cells, followed by a 3-week engraftment period and randomized by weight into 6 groups (Table 1). Groups were treated as follows: untreated control, IV nab-paclitaxel delivered via tail-vein injection: 5.0 mg/kg once weekly for 3 weeks, inhaled NanoPac: 0.5 mg/kg once weekly for 4 weeks; 1.0 mg/kg once weekly for 4 weeks; 0.5 mg/kg twice weekly for 4 weeks, and 1.0 mg/kg twice weekly for 4 weeks. Nebulized NanoPac was delivered via 2 parallel Up-Mist compressed air jet nebulizers into a rodent nose-only inhalation exposure chamber (Figure 4). Necropsy and tissue collection occurred 14 days post final treatment (Group 2), 4 days post final treatment (Groups 3 and 4) and 1 day post final treatment (Groups 5 and 6). Histology was performed with hematoxylin & eosin (H&E) (each group n=20); immunohistochemistry using AE1/AE3+ and CD11b+ (Group 1 n=2; Groups 2-6 n=3) and BCL6+ (Groups 1, 2 and 3 n=1).

Results: Tumor regression was identified in H&E stained slides by scalloping of tumor nodule edges, partial to complete tumor cell loss, the presence of fibrous connective tissue scaffolding in the parenchyma and foamy macrophage infiltration. Inhaled NanoPac treated rats had statistically greater incidence of tumor regression compared to untreated controls and IV nab-paclitaxel treated animals (Figure 7). Immunohistochemistry revealed the presence and intensity of tumor regression and immune response. Untreated control: robust tumor cell growth (2/2), mild lymphoid infiltrates and 0.5-1/low-power microscopy field (LPMF) tertiary lymphoid structures (TLS). IV nab-paclitaxel: some tumor regression (2/3), mild and moderate lymphoid infiltrates and approximately 1 TLS/LPMF. Inhaled NanoPac: some degree of tumor regression (12/12, with 2 showing complete regression), moderate and marked lymphoid infiltrates with 2-3 TLS/LPMF (Table 2).

Figure 1. Submicron Particle Production Technology

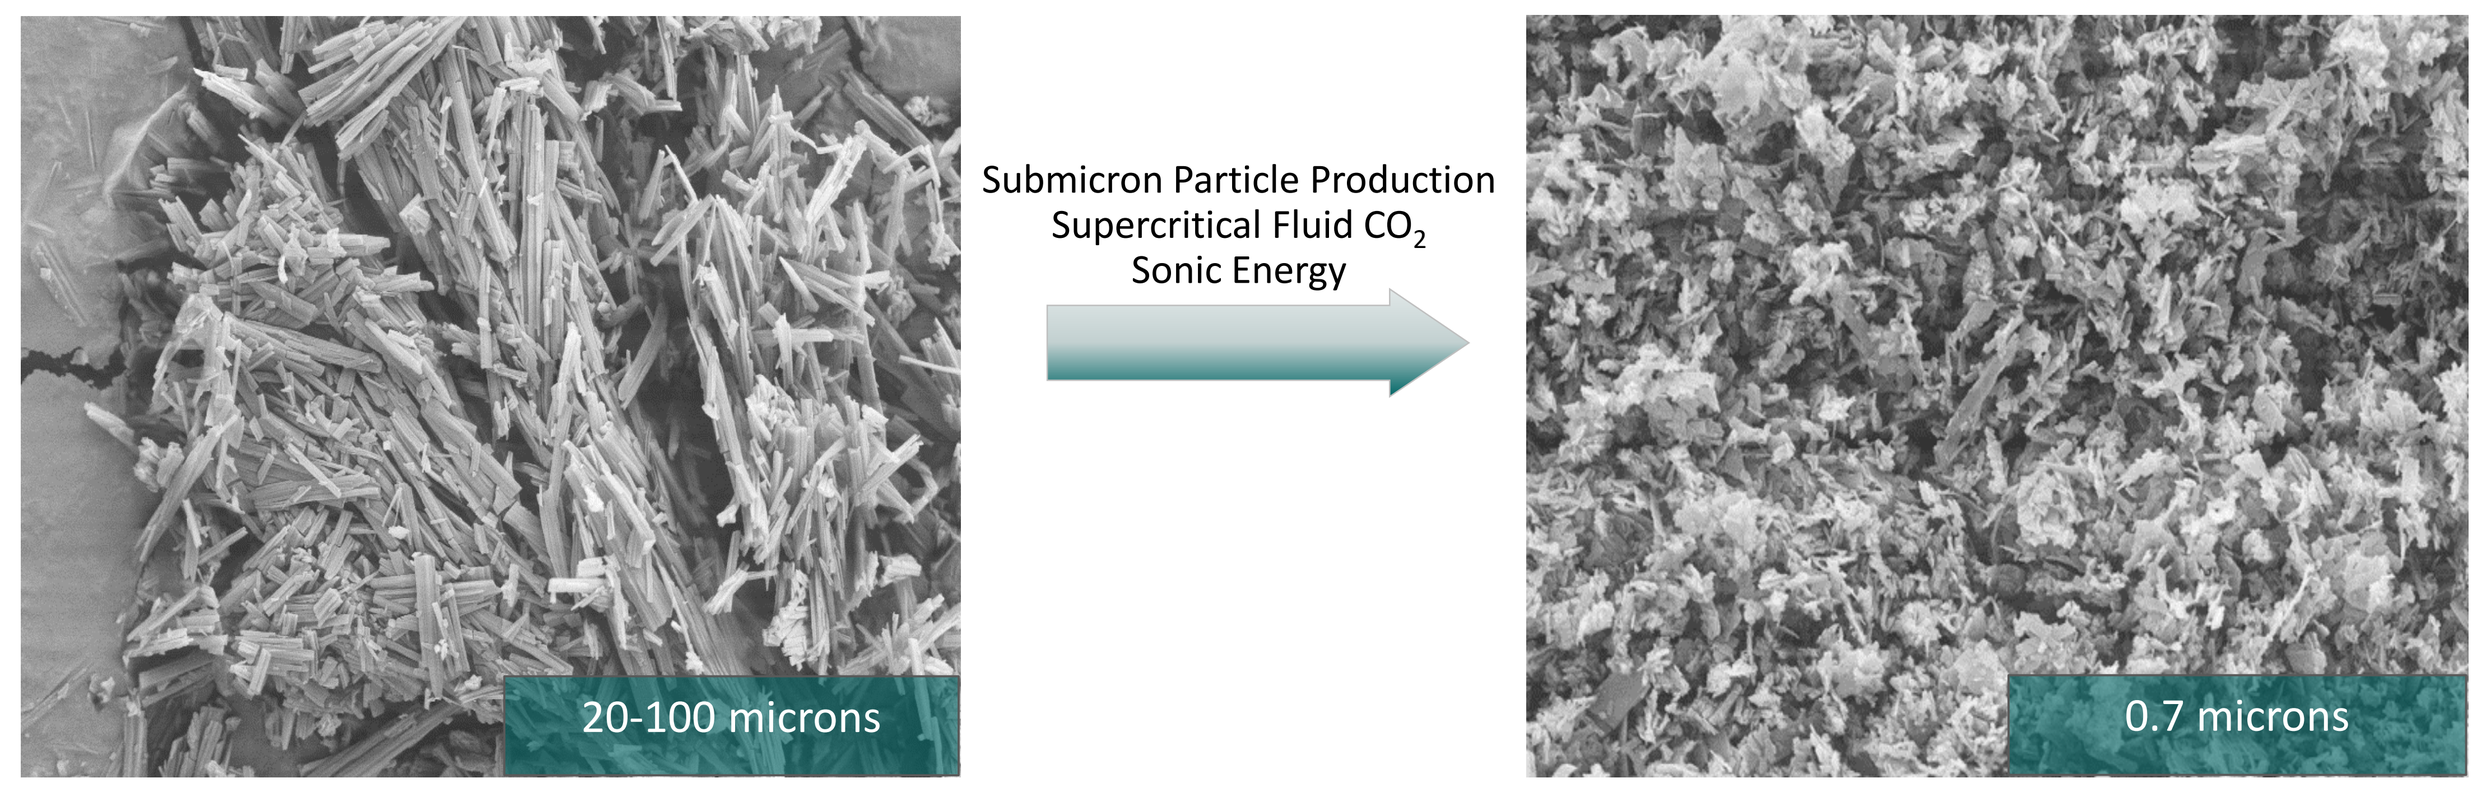
20-100 microns
Submicron Particle Production
Supercritical Fluid CO_2
Sonic Energy
0.7 microns

CritiTech, Inc.; Lawrence, KS

- Large active pharmaceutical ingredient (API) crystals cannot be suspended and must be dissolved for IV or inhaled delivery
- Small amount of systemically delivered dose reaches the tumor site for a short period of time[1]

CritiTech, Inc.; Lawrence, KS

- Each submicron particle contains 2-3 billion molecules of paclitaxel
- Increased particle surface area allows for sustained therapeutic API release
- Suspended particles can be nebulized for local pulmonary delivery

Figure 2. Lung Tissue Paclitaxel Concentration Time Curve and PK Data

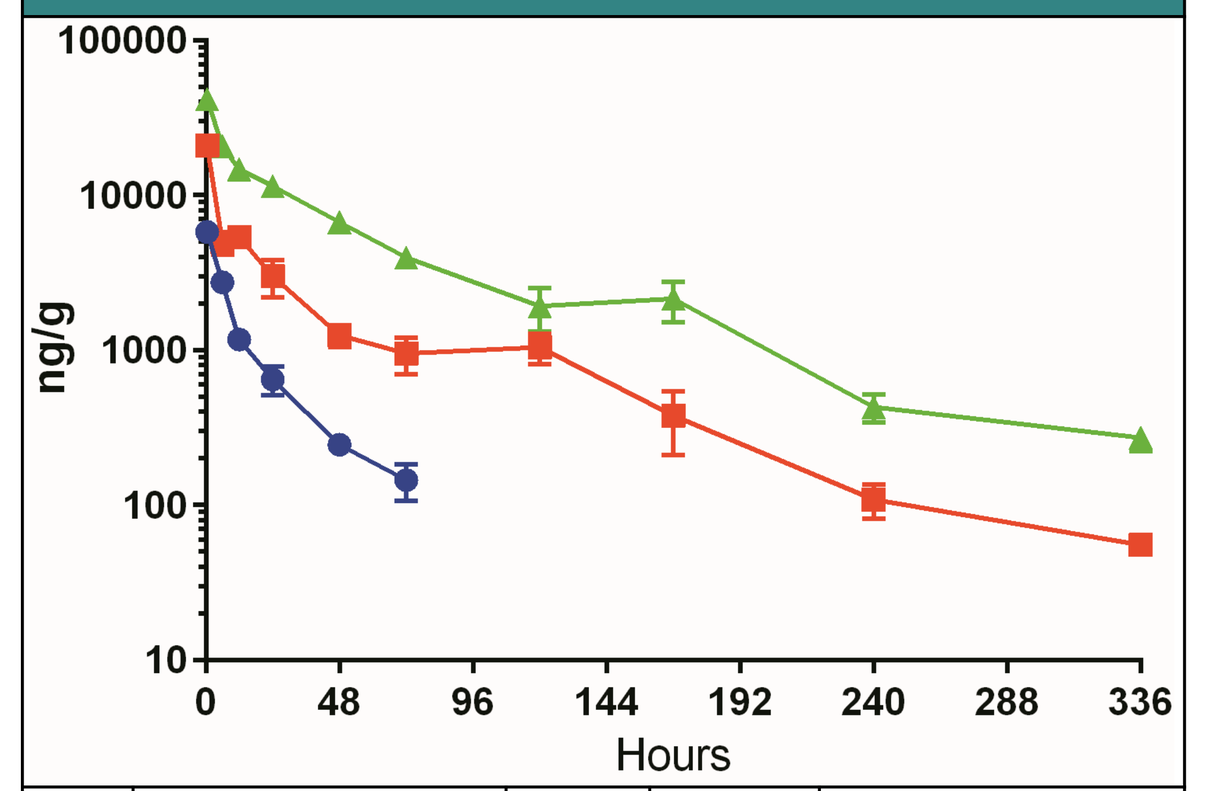
ng/g
Hours

	Group Dose (mg/kg)	$T_{1/2}$ (hours)	C_{max} (ng/g)	$AUC_{D(last)}$ (hr*ng*mg/g*kg)
▲	Inhaled NanoPac (1.18)	56.0	41,600	997,985
■	Inhaled NanoPac (0.38)	56.3	21,000	914,095
●	IV nab-paclitaxel (2.9)	19.9	5,800	23,112

Figure 3. Plasma Paclitaxel Concentration Time Curve and PK Data

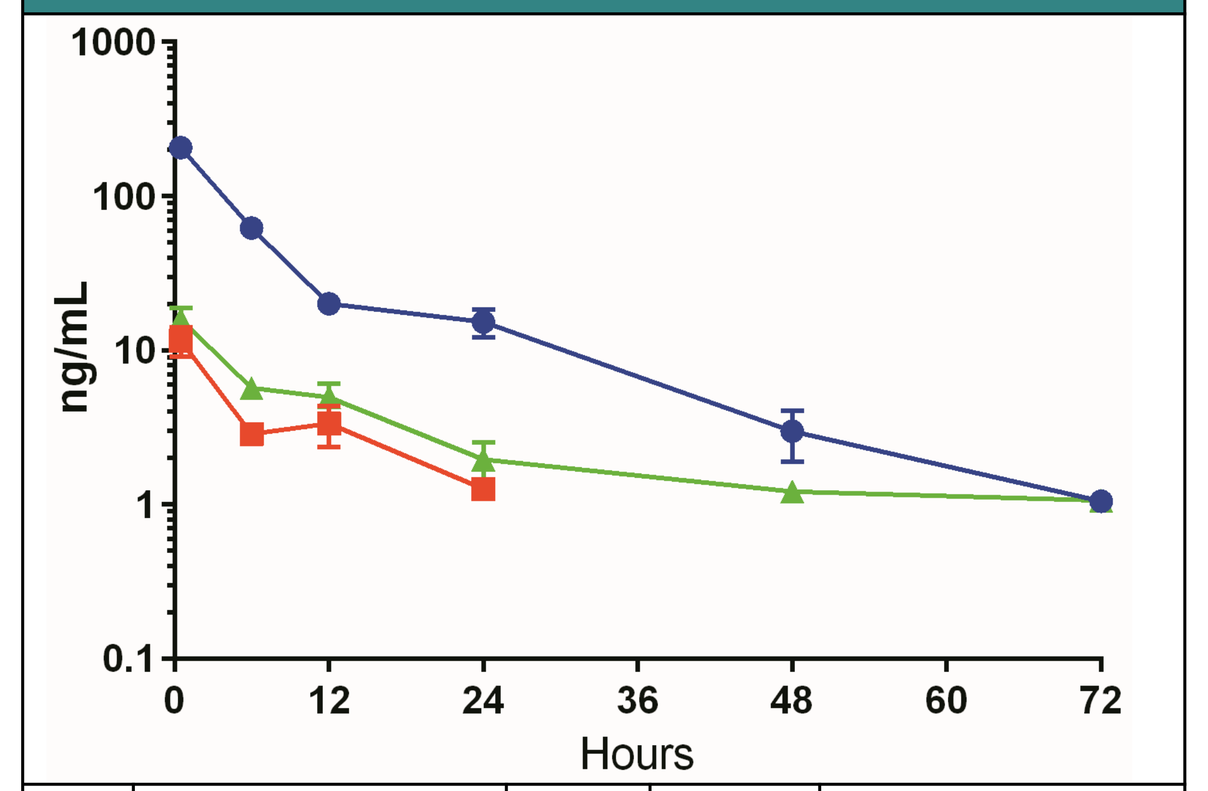
ng/mL
Hours

	Group Dose (mg/kg)	$T_{1/2}$ (hours)	C_{max} (ng/mL)	$AUC_{D(last)}$ (hr*ng*mg/mL*kg)
▲	Inhaled NanoPac (1.18)	8.3	15.9	193
■	Inhaled NanoPac (0.38)	7.9	11.6	264
●	IV nab-paclitaxel (2.9)	8.7	206	528

To evaluate the potential for local delivery of NanoPac to the lungs via inhalation, a preclinical pharmacokinetic study was first conducted to confirm prolonged paclitaxel residence in the lung when administered via nebulized inhalation in healthy male Sprague-Dawley rats in one of two inhaled doses (0.38 or 1.18 mg/kg) compared to intravenous administration of nab-paclitaxel (2.9 mg/kg)[1].

Table 1. Study Design (n = 20/Group)

Group	Treatment	Frequency
1	Control (no treatment)	N/A
2	IV nab-paclitaxel; 5.0 mg/kg	Once weekly for 3 weeks
3	Inhaled NanoPac; 0.5 mg/kg	Once weekly for 4 weeks
4	Inhaled NanoPac; 1.0 mg/kg	Once weekly for 4 weeks
5	Inhaled NanoPac; 0.5 mg/kg	Twice weekly for 4 weeks
6	Inhaled NanoPac; 1.0 mg/kg	Twice weekly for 4 weeks

120 x-irradiated nude rats were intratracheally instilled with 20 x 10^6 Calu-3 cancer cells; followed by a 3-week engraftment period, and randomized by weight into one of six groups.

Figure 4. Nose-Only Inhalation Chamber

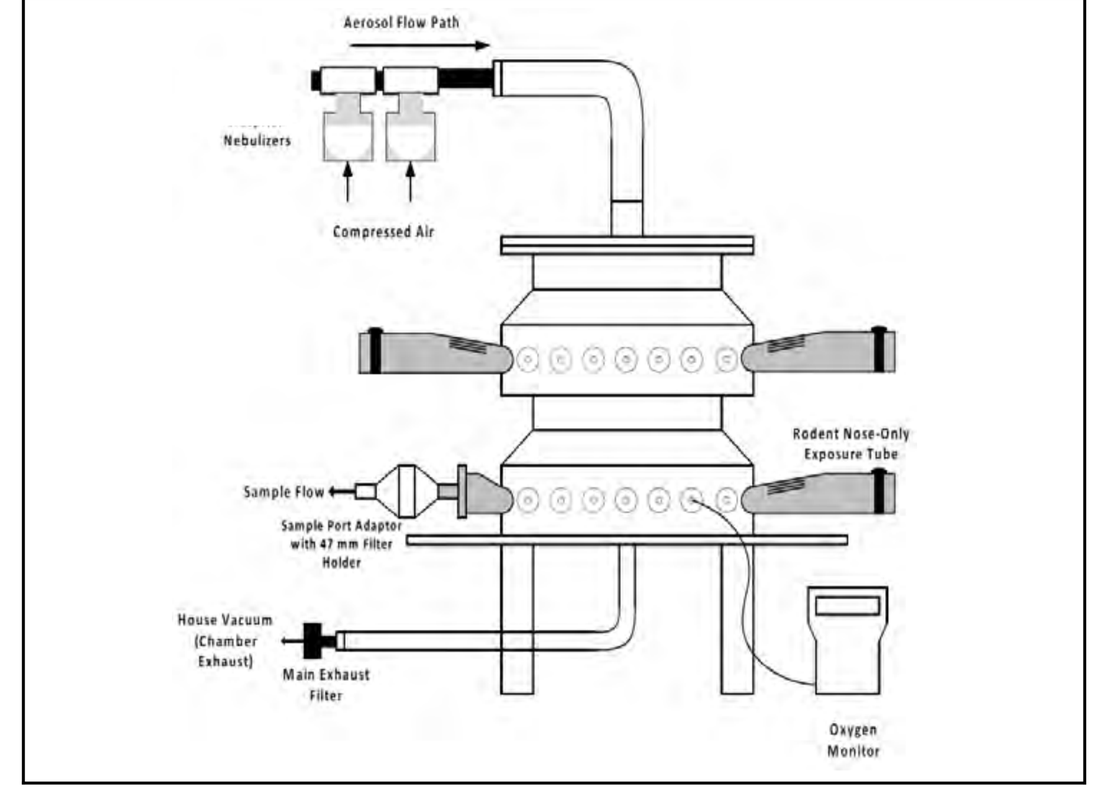
Aerosol Flow Path
Nebulizers
Compressed Air
Rodent Nose-Only Exposure Tube
Sample Flow
Sample Port Adaptor with 47 mm Filter Holder
House Vacuum (Chamber Exhaust)
Main Exhaust Filter
Oxygen Monitor

Inhaled NanoPac was delivered via 2-parallel Up-Mist compressed air jet nebulizers into a rodent Nose-only exposure chamber.

NanoPac is a registered trademark of NanOlogy, LLC.
Up-Mist is a registered trademark of UnoMedical A/S.

Figure 5. Group 1 H&E and Immunohistochemistry Staining

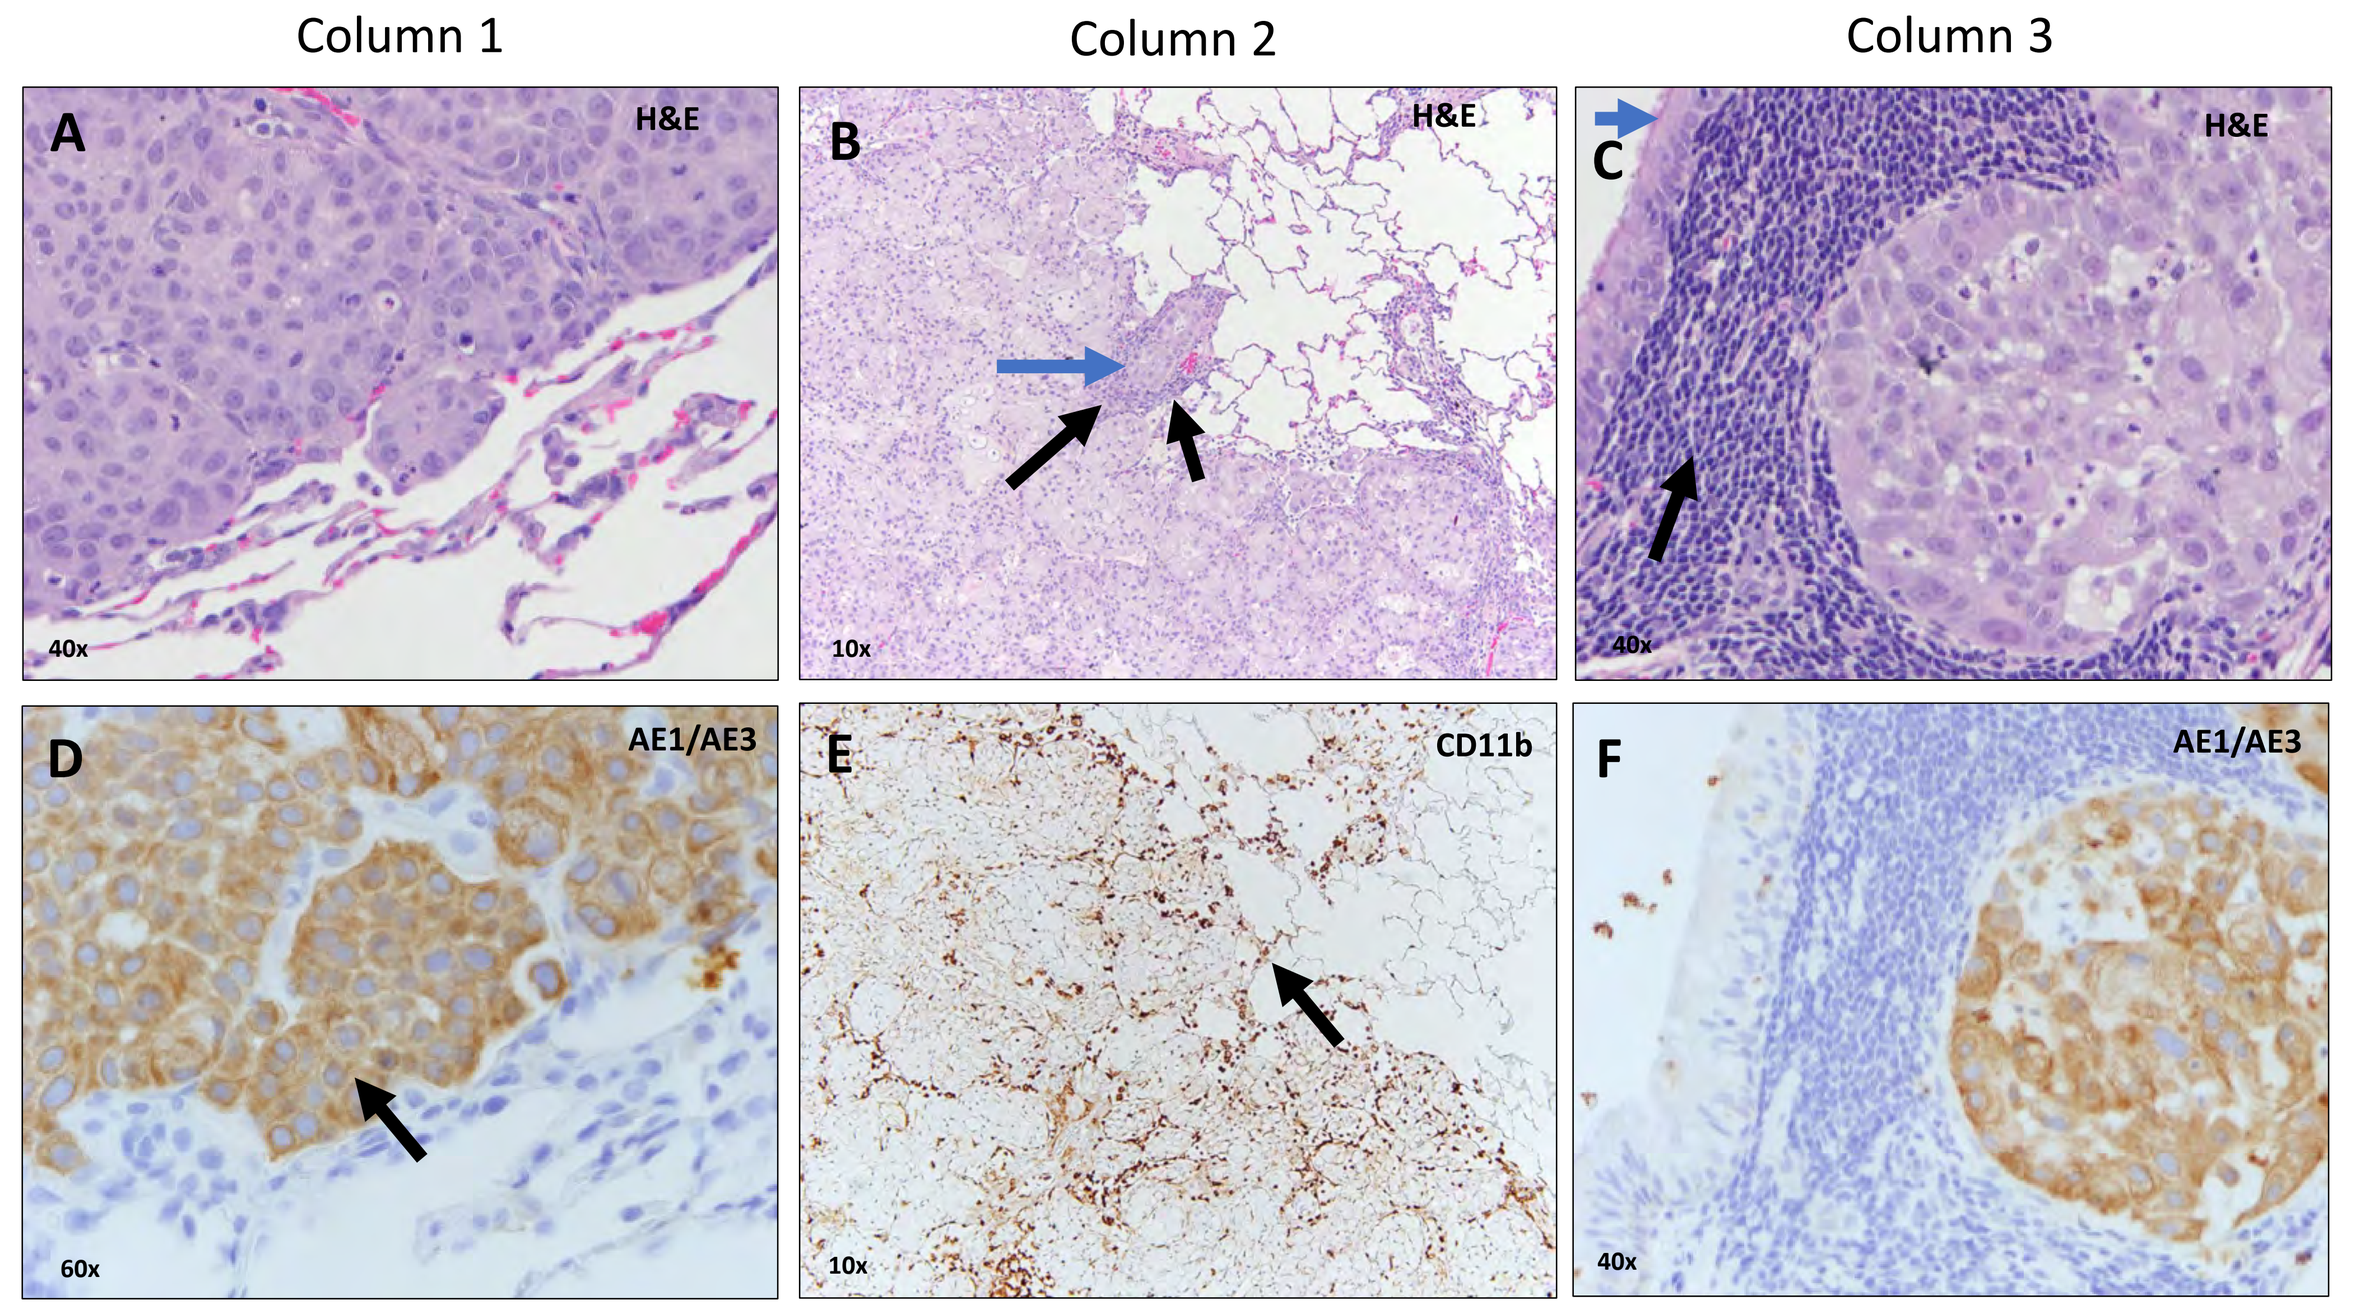
Column 1
Column 2
Column 3
A H&E 40x
B H&E 10x
C H&E 40x
D AE1/AE3 60x
E CD11b 10x
F AE1/AE3 40x

Column 1: H&E staining of adenocarcinoma (A) and corresponding keratin (AE1/AE3) staining (D) highlighting specific labeling of carcinoma cells (black arrow) in untreated lung tissue.

Column 2: H&E staining of adenocarcinoma (B) showing focal rudimentary duct formation (blue arrow) with a limited immune cell component consisting of lymphocytes and focal macrophages (black arrows). CD11b staining (E) highlighting minimal natural killer cells and macrophages (black arrow).

Column 3: H&E staining of adenocarcinoma (C) next to bronchus-associated lymphoid tissue containing mature lymphocytes (black arrow) and normal bronchial lining (blue arrow top left). Corresponding keratin (AE1/AE3) staining (F) highlighting carcinoma cells and lack of lymphoid cell staining.

Figure 6. Group 2 H&E and Immunohistochemistry Staining

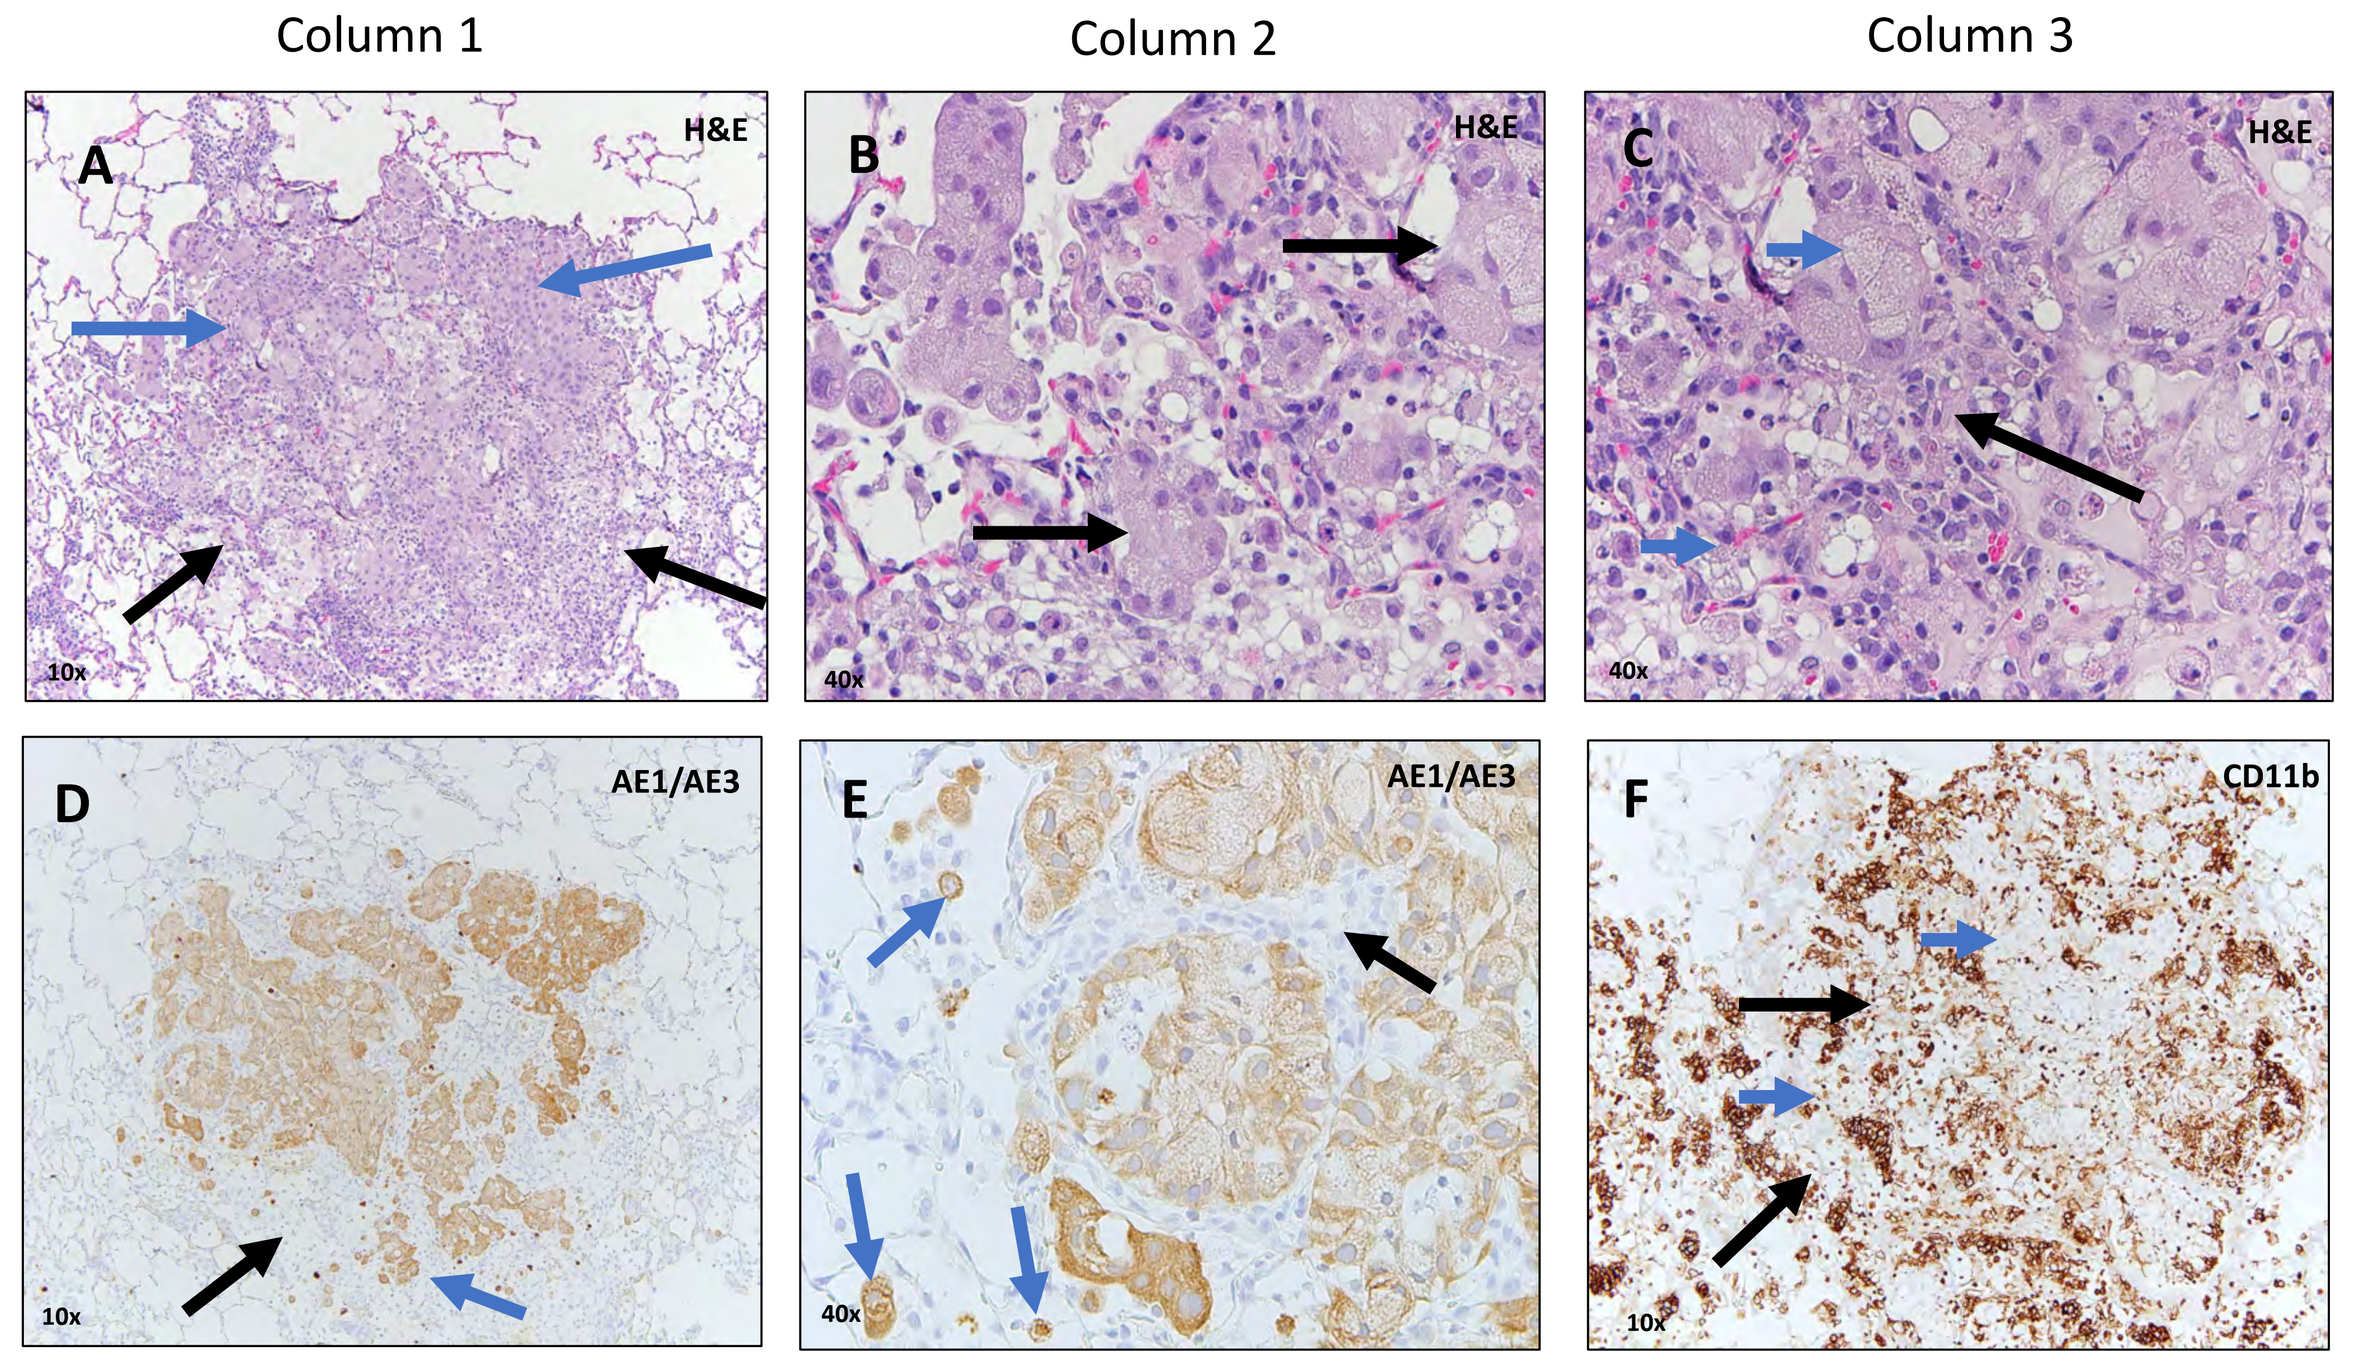
Column 1
Column 2
Column 3
A H&E 10x
B H&E 40x
C H&E 40x
D AE1/AE3 10x
E AE1/AE3 40x
F CD11b 10x

Column 1: (A) H&E staining of adenocarcinoma (blue arrows) showing progressive separation of tumor cells and increased immune cell response (black arrows). Corresponding keratin (AE1/AE3) staining (D) showing tumor cluster separation (blue arrow) and intervening stroma (black arrow).

Column 2: Higher magnification view (B) of image A showing smaller clusters of tumor cells (black arrows). Keratin (AE1/AE3) staining (E) higher magnification view of image D showing separated tumor cell nodules, decreasing tumor cell clusters, individual single tumor cells (blue arrows), and unstained intervening stroma containing immune cells (black arrow).

Column 3: H&E staining (C) showing immune cells (black arrow) in center of tumor nodule (blue arrows). Corresponding low magnification view of a CD11b-stain (F) of image A showing increased density of immune cells (black arrows) within the tumor cell clusters and residual carcinoma not labeled with CD11b (blue arrows).

Figure 7. Animals Exhibiting Tumor Regression

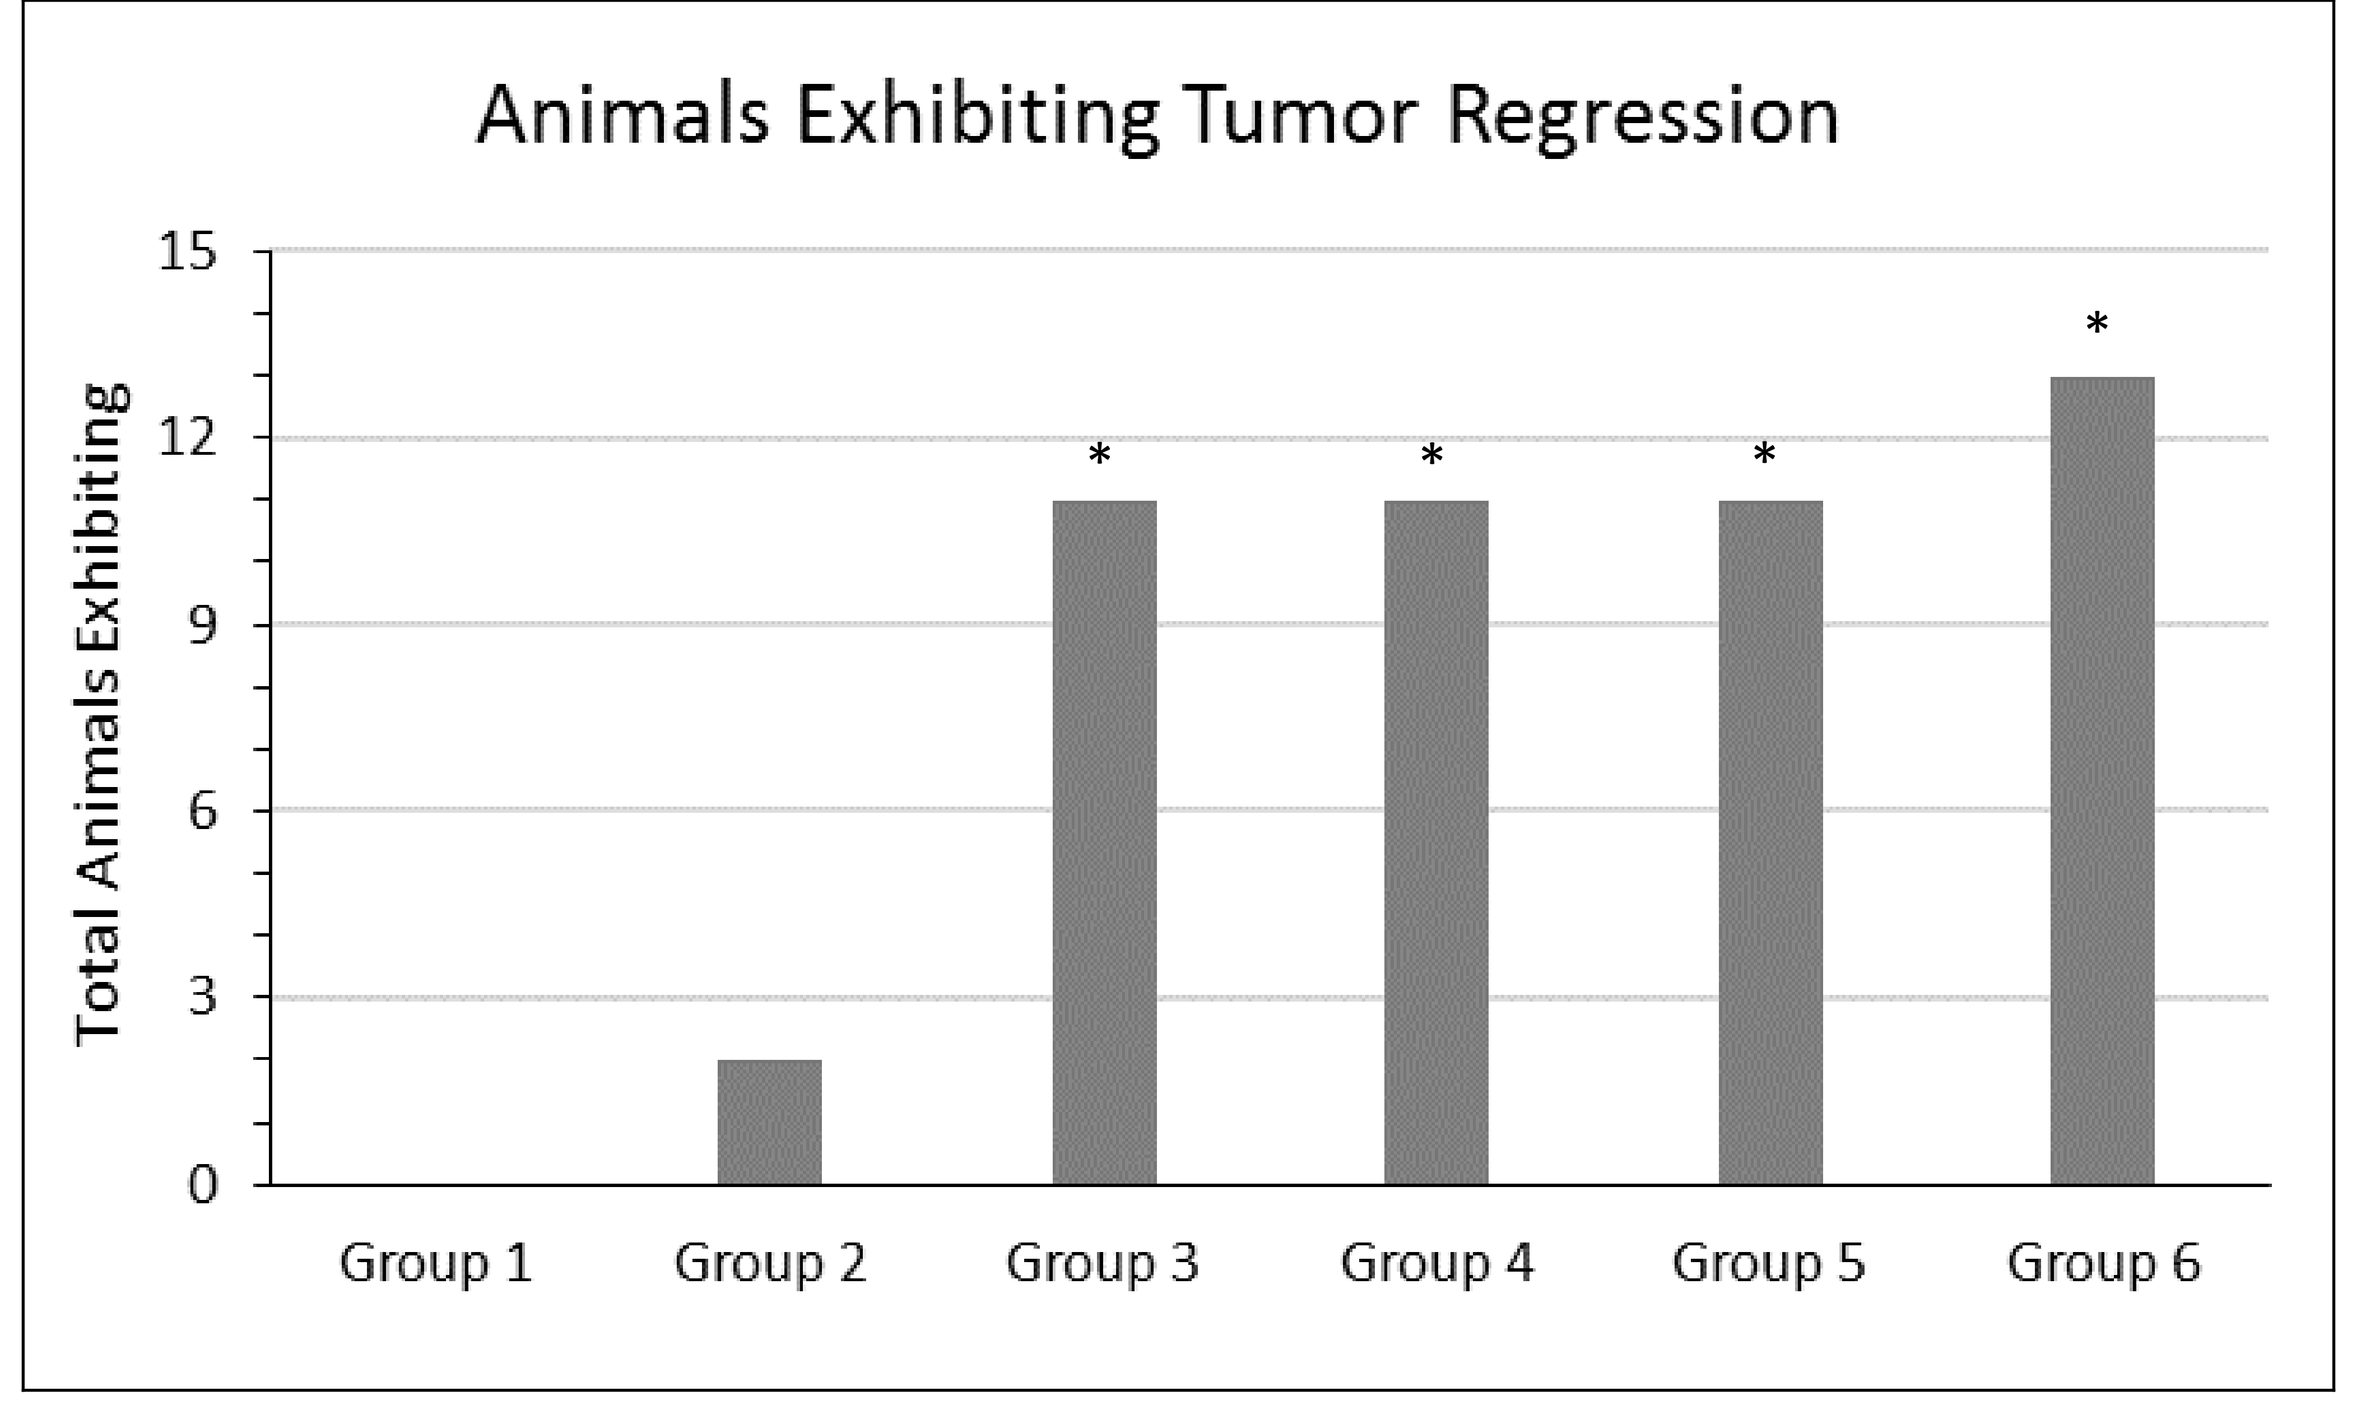
Animals Exhibiting Tumor Regression
Total Animals Exhibiting
0
3
6
9
12
15
Group 1
Group 2
Group 3
Group 4
Group 5
Group 6
*

Tumor regression identified via H&E stained slides by scalloping of tumor nodule edges, partial to complete tumor cell loss, presence of fibrous connective tissue scaffolding in the parenchyma and foamy macrophage infiltration. Rats treated with inhaled NanoPac had statistically greater incidence of tumor regression compared to untreated and nab-paclitaxel treated rats (p<0.01; two-tailed chi-squared*).

Figure 8. Groups 3 and 4 H&E and Immunohistochemistry Staining

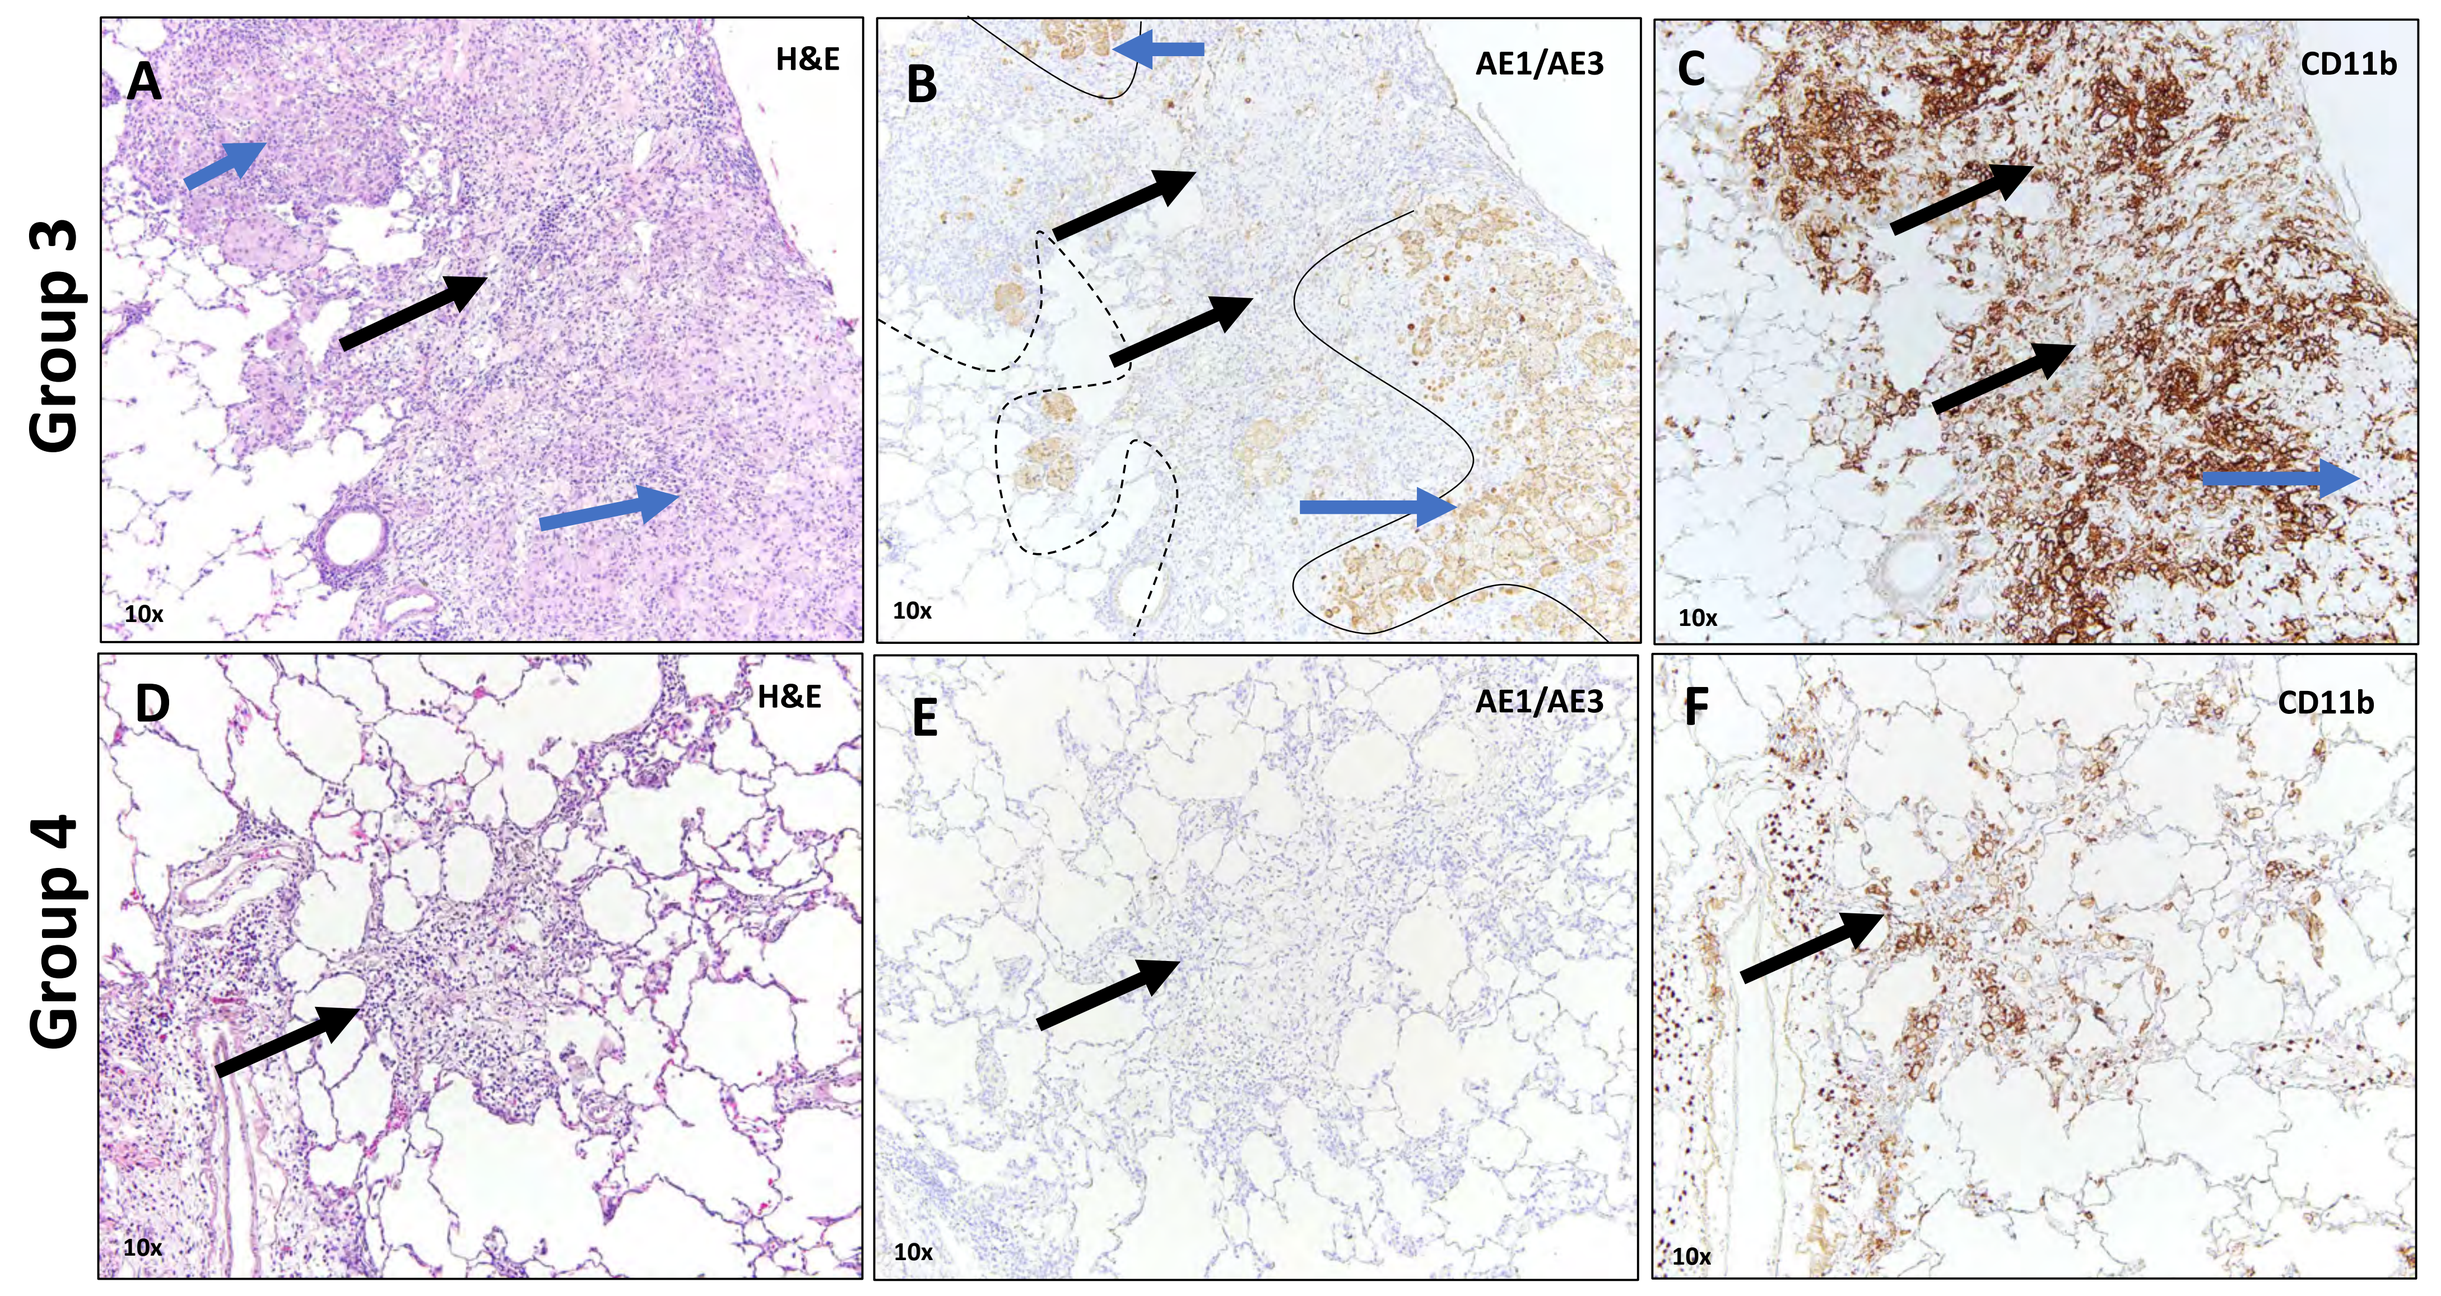
Group 3
Group 4
A H&E 10x
B AE1/AE3 10x
C CD11b 10x
D H&E 10x
E AE1/AE3 10x
F CD11b 10x

Group 3: H&E staining of adenocarcinoma (A) showing regression highlighted by separation and loss of tumor cells at tumor periphery (blue arrows) and non-neoplastic stroma with inflammation separating carcinoma into nodules (black arrow). Keratin (AE1/AE3) staining (B) showing residual carcinoma (blue arrows), original carcinoma border (dashed black line) and residual carcinoma border (continuous black lines); unstained areas (black arrows) represents area of tumor loss. CD11b staining (C) showing immune cell infiltration in areas of tumor regression (black arrows) and unstained residual carcinoma (blue arrow).

Group 4: H&E staining (D) showing no viable adenocarcinoma: complete regression, with organizing inflammation composed of fibrous stroma with admixed lymphocytes and macrophages (black arrow). Lack of positive keratin (AE1/AE3) staining (E) in the same area as image D confirms morphologic absence of residual carcinoma (black arrow), and CD11b stain (F) shows mild to moderate immune cell infiltrate (black arrow).

Figure 9. Groups 5 and 6 H&E and Immunohistochemistry Staining

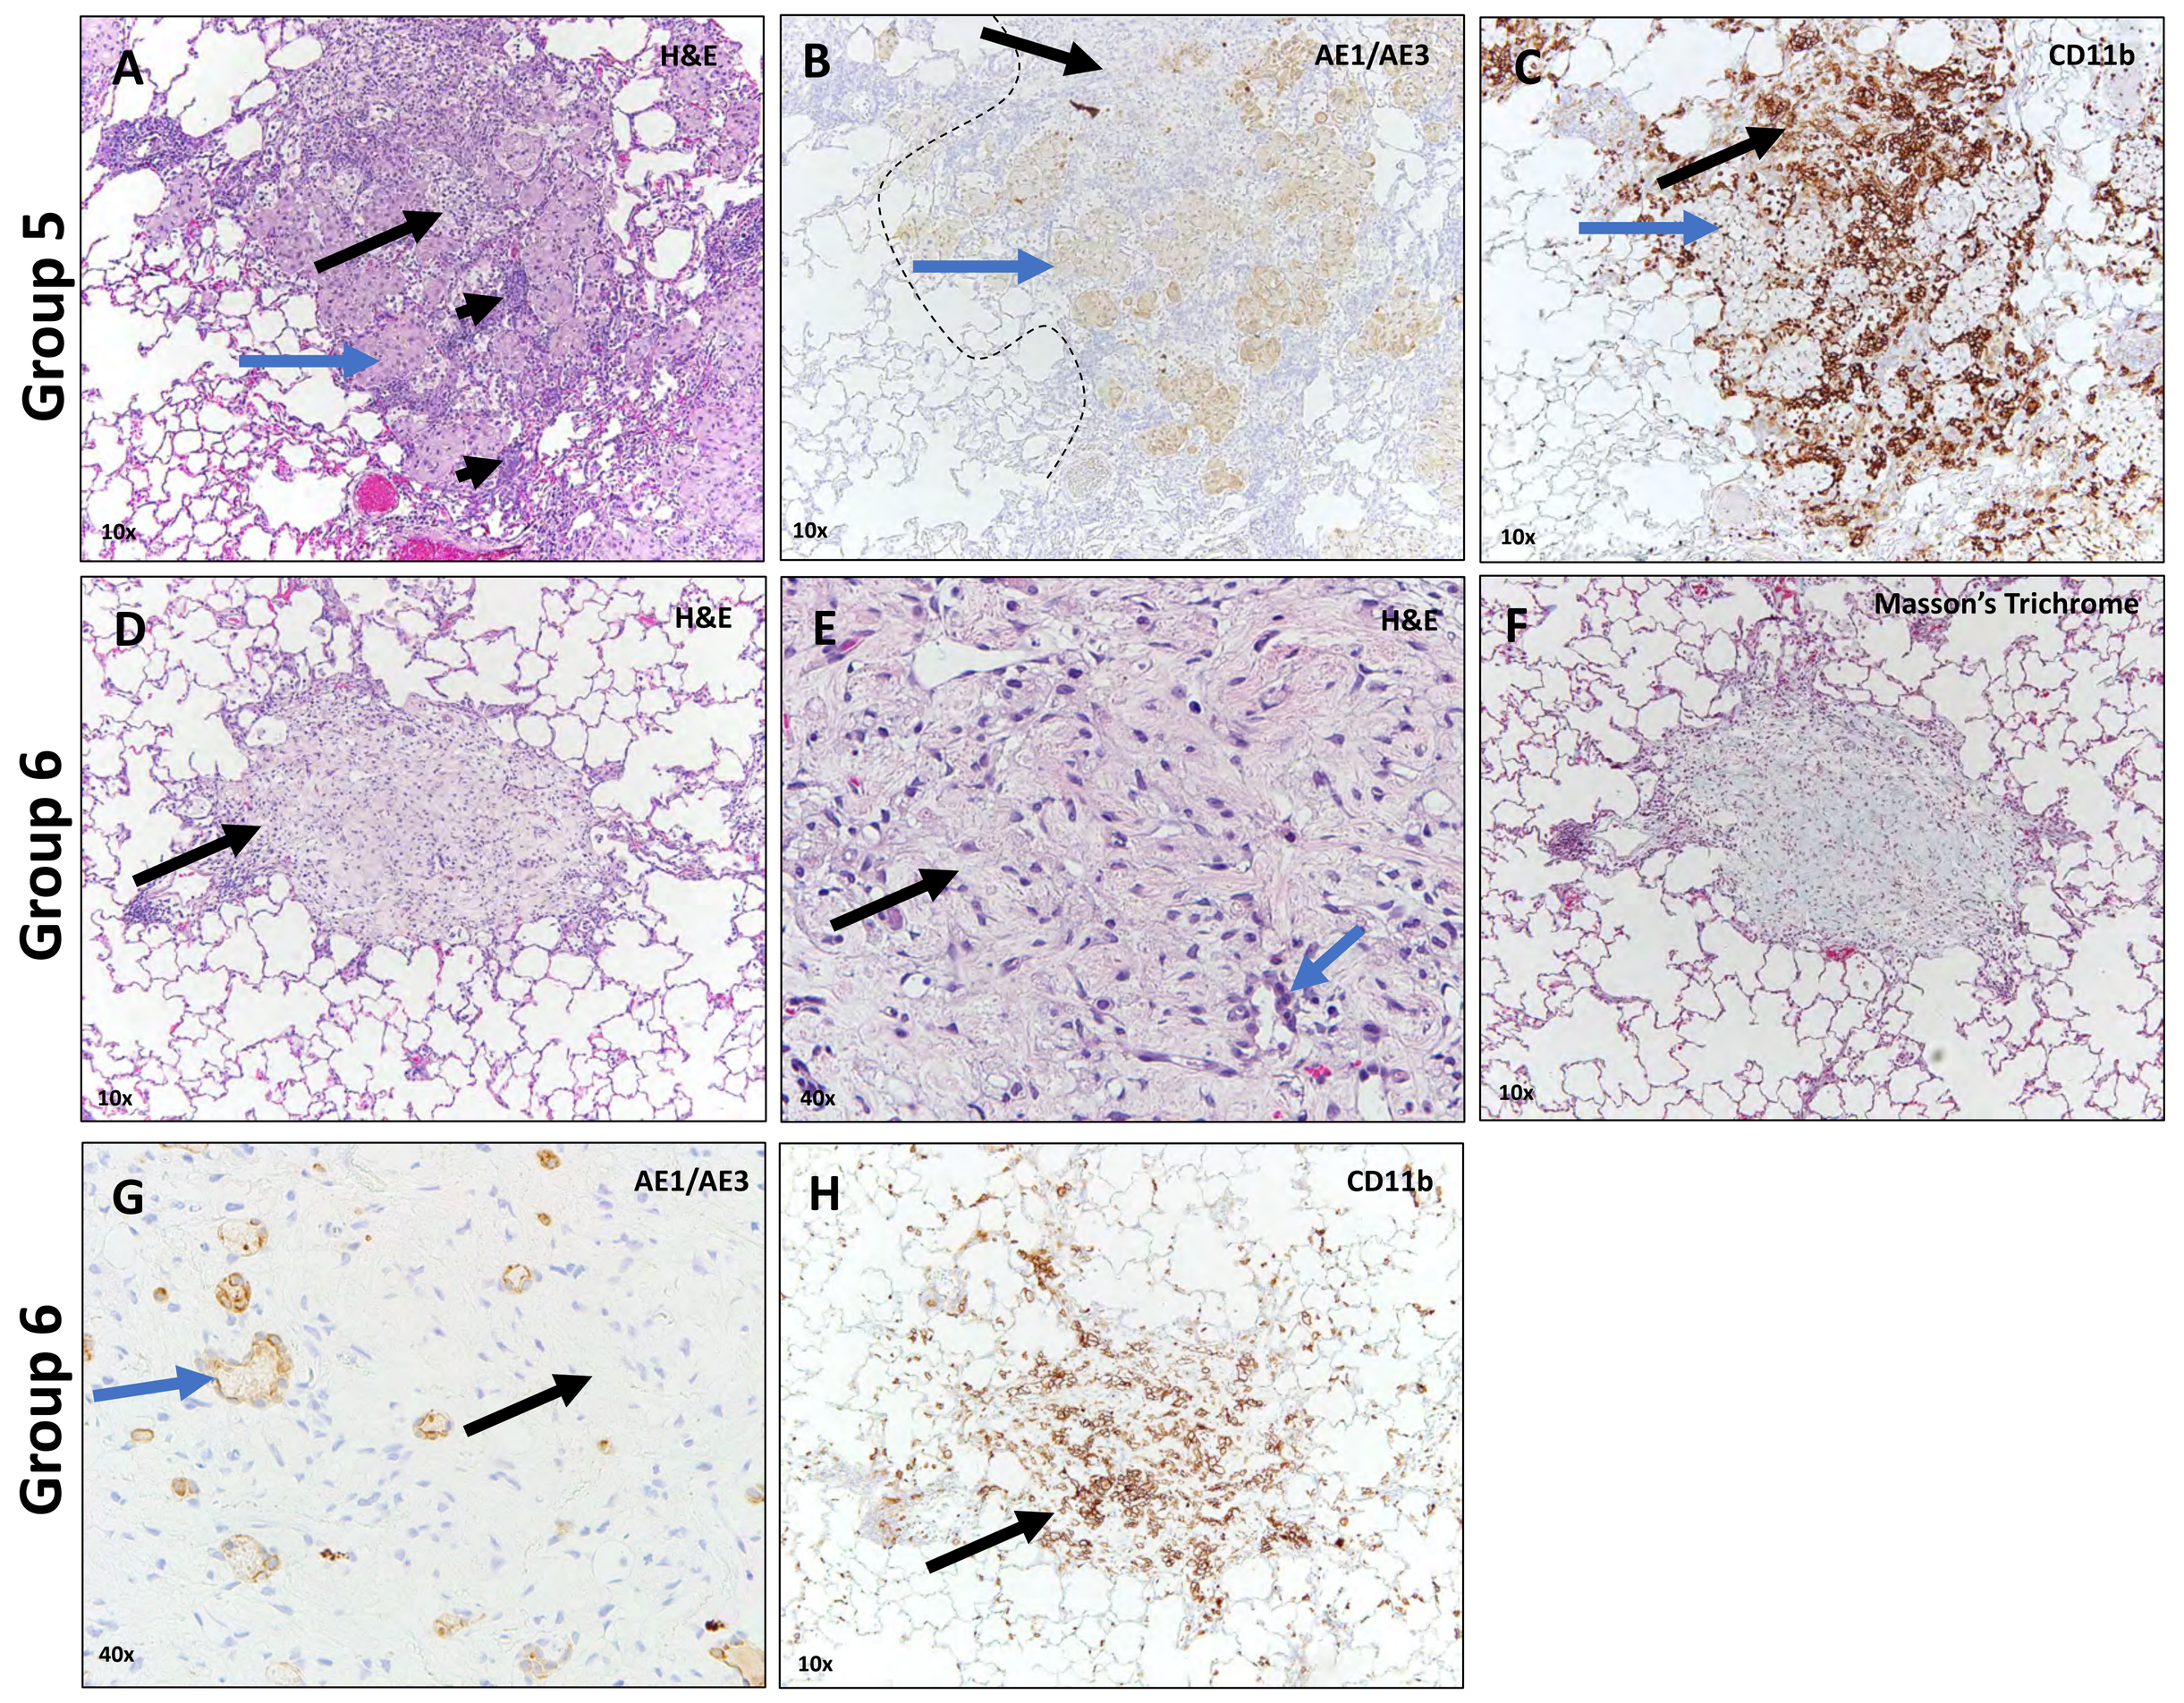
Group 5
Group 6
Group 6
A H&E 10x
B AE1/AE3 10x
C CD11b 10x
D H&E 10x
E H&E 40x
F Masson's Trichrome 10x
G AE1/AE3 40x
H CD11b 10x

Group 5: H&E staining of adenocarcinoma (A) showing regression within a tumor nodule (blue arrow), increased intra-nodular stroma (long black arrow) and increased intra- and peri-nodular lymphoid cells (short black arrows). Keratin (AE1/AE3) staining (B) showing carcinoma (blue arrow), large unstained area of tumor loss (black arrow) and original carcinoma border (dashed black line). CD11b staining (C) showing immune cell infiltrate in area of regression (black arrow) and surrounding areas of unstained carcinoma (blue arrow).

Group 6: H&E staining showing cluster of foamy cells (black arrow) (D), and increased magnification (E) of image D highlighting cells with foamy cytoplasm (black arrow) and regenerating small blood vessels or alveoli (blue arrow). Masson's trichrome (F) showing blue-stained collagen (fibrous organization). Keratin (AE1/AE3) staining (G) labeling single cells and duct-like structures (blue arrow) representing a combination of regenerative or atrophic non-neoplastic structures such as small blood vessels or alveoli as well as unstained intervening foamy cells (black arrow). CD11b staining (H) corresponding to image D shows immune cell infiltrate in area of tumor regression (black arrow).

Table 2. Summary Immunohistopathology Scoring for Tumor Regression

Group Number	N	Regression: 0% of nodules	Regression: 1-10% of nodules	Regression: 11-50% of nodules	Regression: >50% of nodules	Regression: Complete regression	CD11b⁺ Immune Cell Infiltration: Mild	CD11b⁺ Immune Cell Infiltration: Moderate	CD11b⁺ Immune Cell Infiltration: Marked	TLS per Low Power Field
1	2	2					2			0.5-1
2	3	1	1		1		1	2		1
3	3		1		2			1	2	3
4	3		1		1	1		2	1	2
5	3				3			2	1	2
6	3				2	1*		2	1	2

Immune cell infiltrate and incidences of tertiary lymphoid structures (measured at 40x power field).
* Residual keratin positive structures noted in one case (rare carcinoma cells versus regenerative or atrophic entrapped blood vessels or alveoli).

Figure 10. Lymphoid Structures

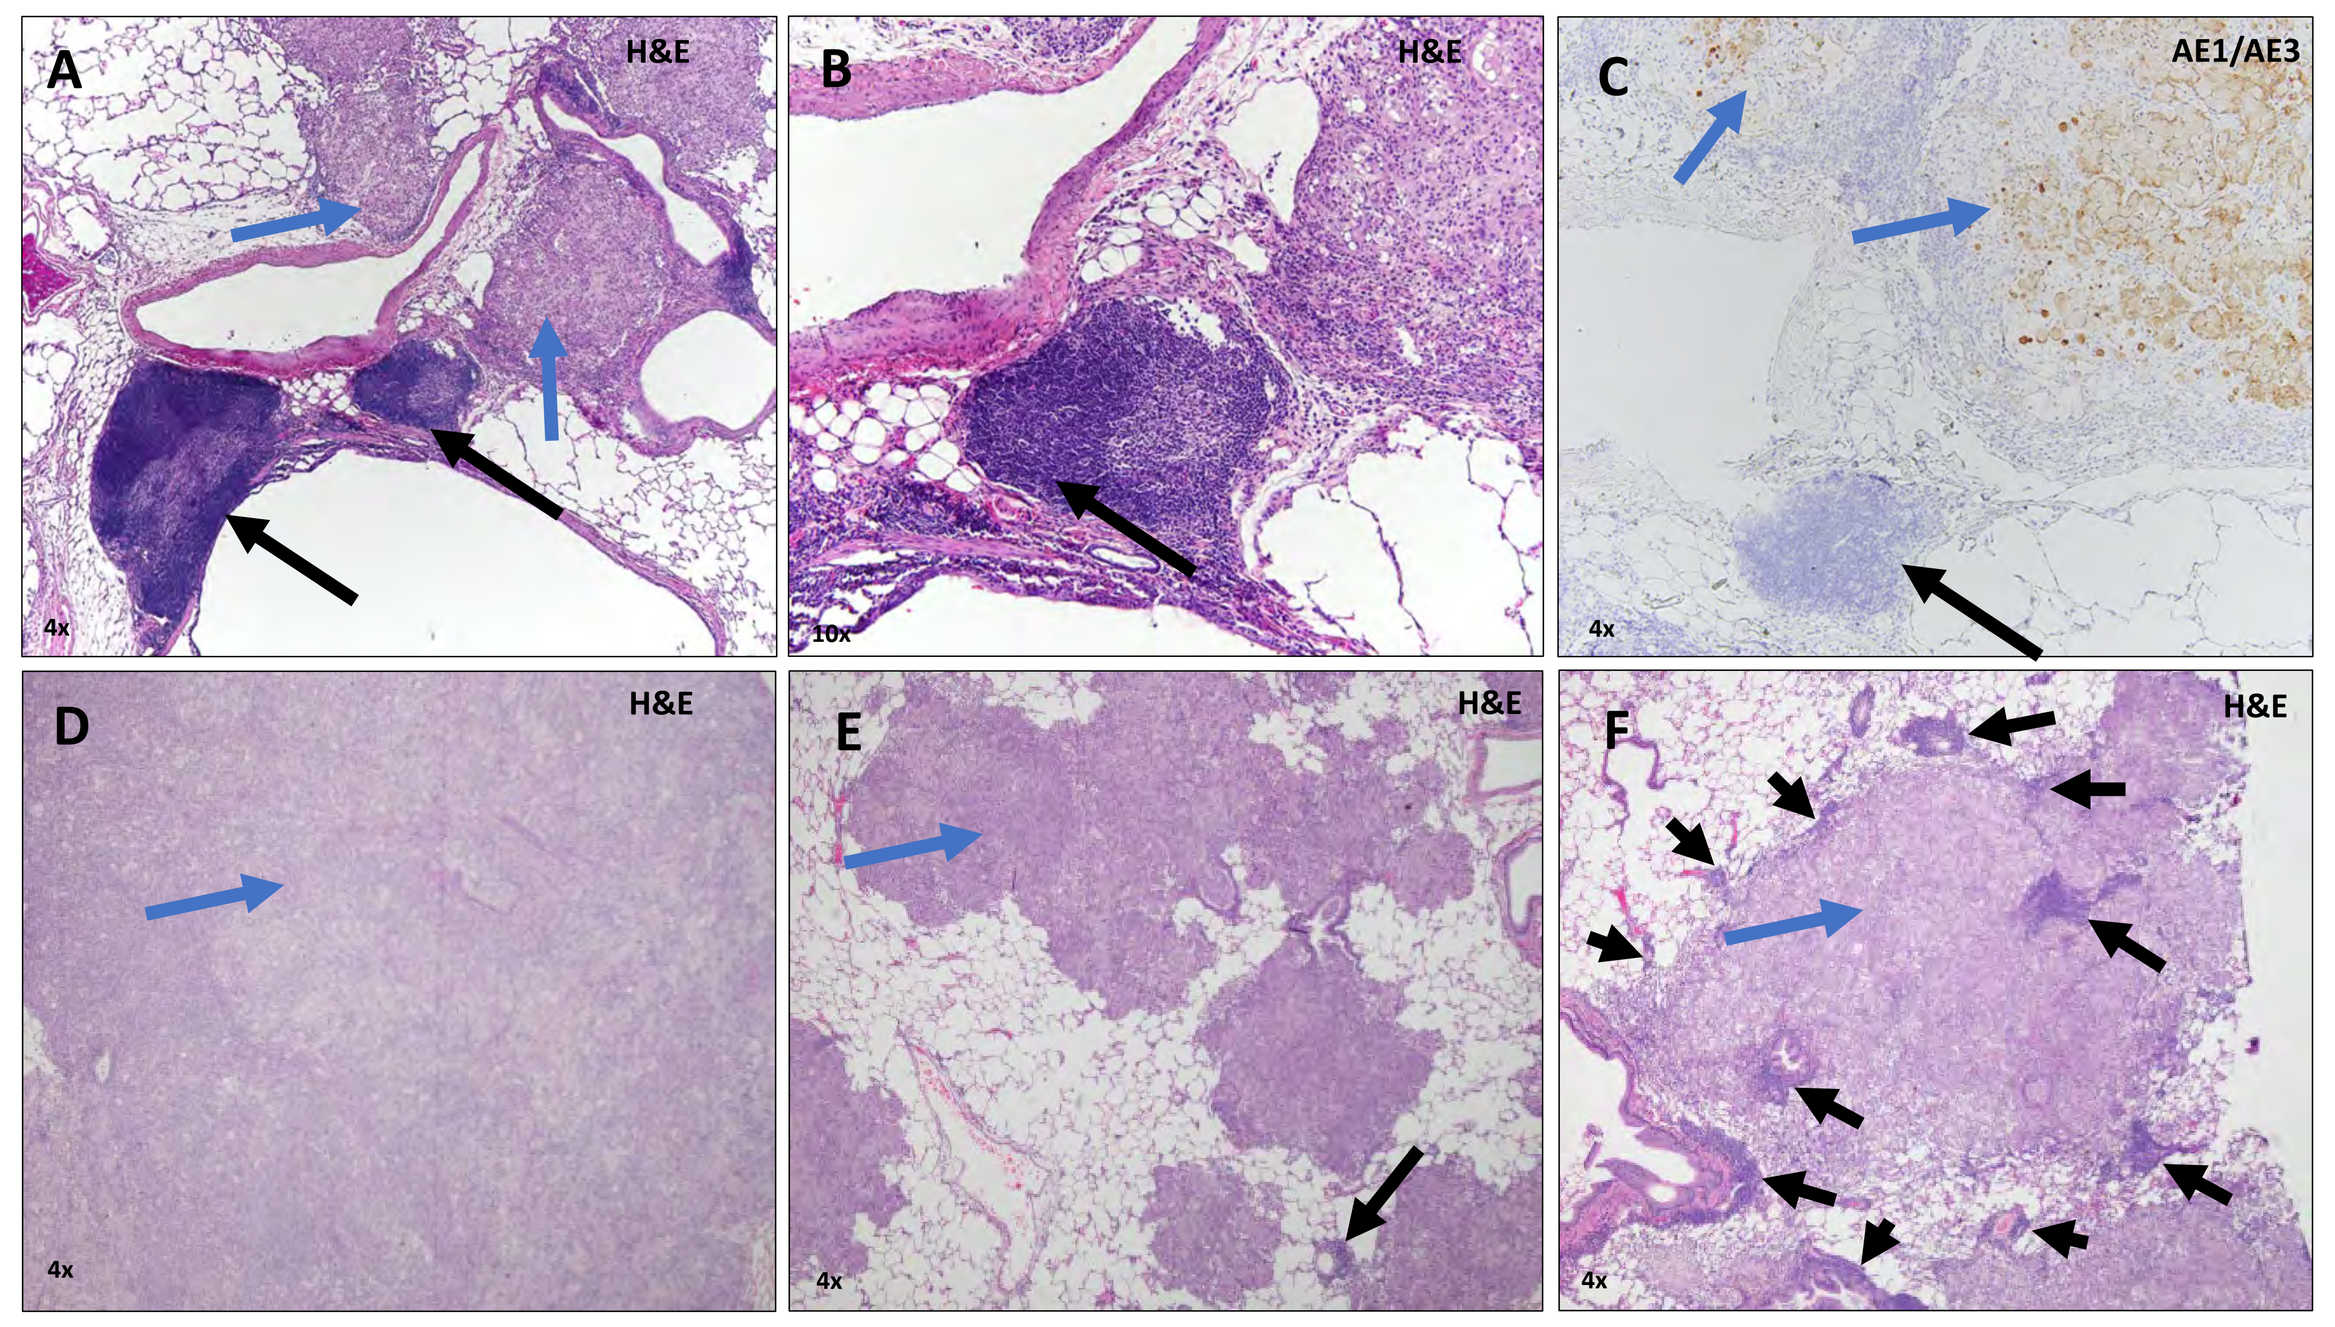
A H&E 4x
B H&E 10x
C AE1/AE3 4x
D H&E 4x
E H&E 4x
F H&E 4x

Top row: Group 6 H&E staining (A) showing two lymphoid structures (black arrows) with distinct lymphoid follicles, germinal centers, paracortical areas and sinuses with adjacent areas of tumor regression (blue arrows). Higher magnification (B) of one of the lymphoid structures from A highlighting a lymphoid follicle with germinal center (black arrow). Keratin (AE1/AE3) staining (C) showing carcinoma (blue arrows) with features of regression, and adjacent unstained lymphoid structure (black arrow).

Bottom row: Group 1 H&E stain (D) showing densely packed adenocarcinoma (blue arrow) and absence of discrete lymphoid collections. Group 2 H&E stain (E) showing adenocarcinoma (blue arrow) with a small collection of lymphoid cells (black arrow). Group 4 H&E stain (F) showing adenocarcinoma (blue arrow) with numerous intra- and peri-tumoral collections of small lymphoid cells (black arrows).

Figure 11. Group 6 Tertiary Lymphoid Structures

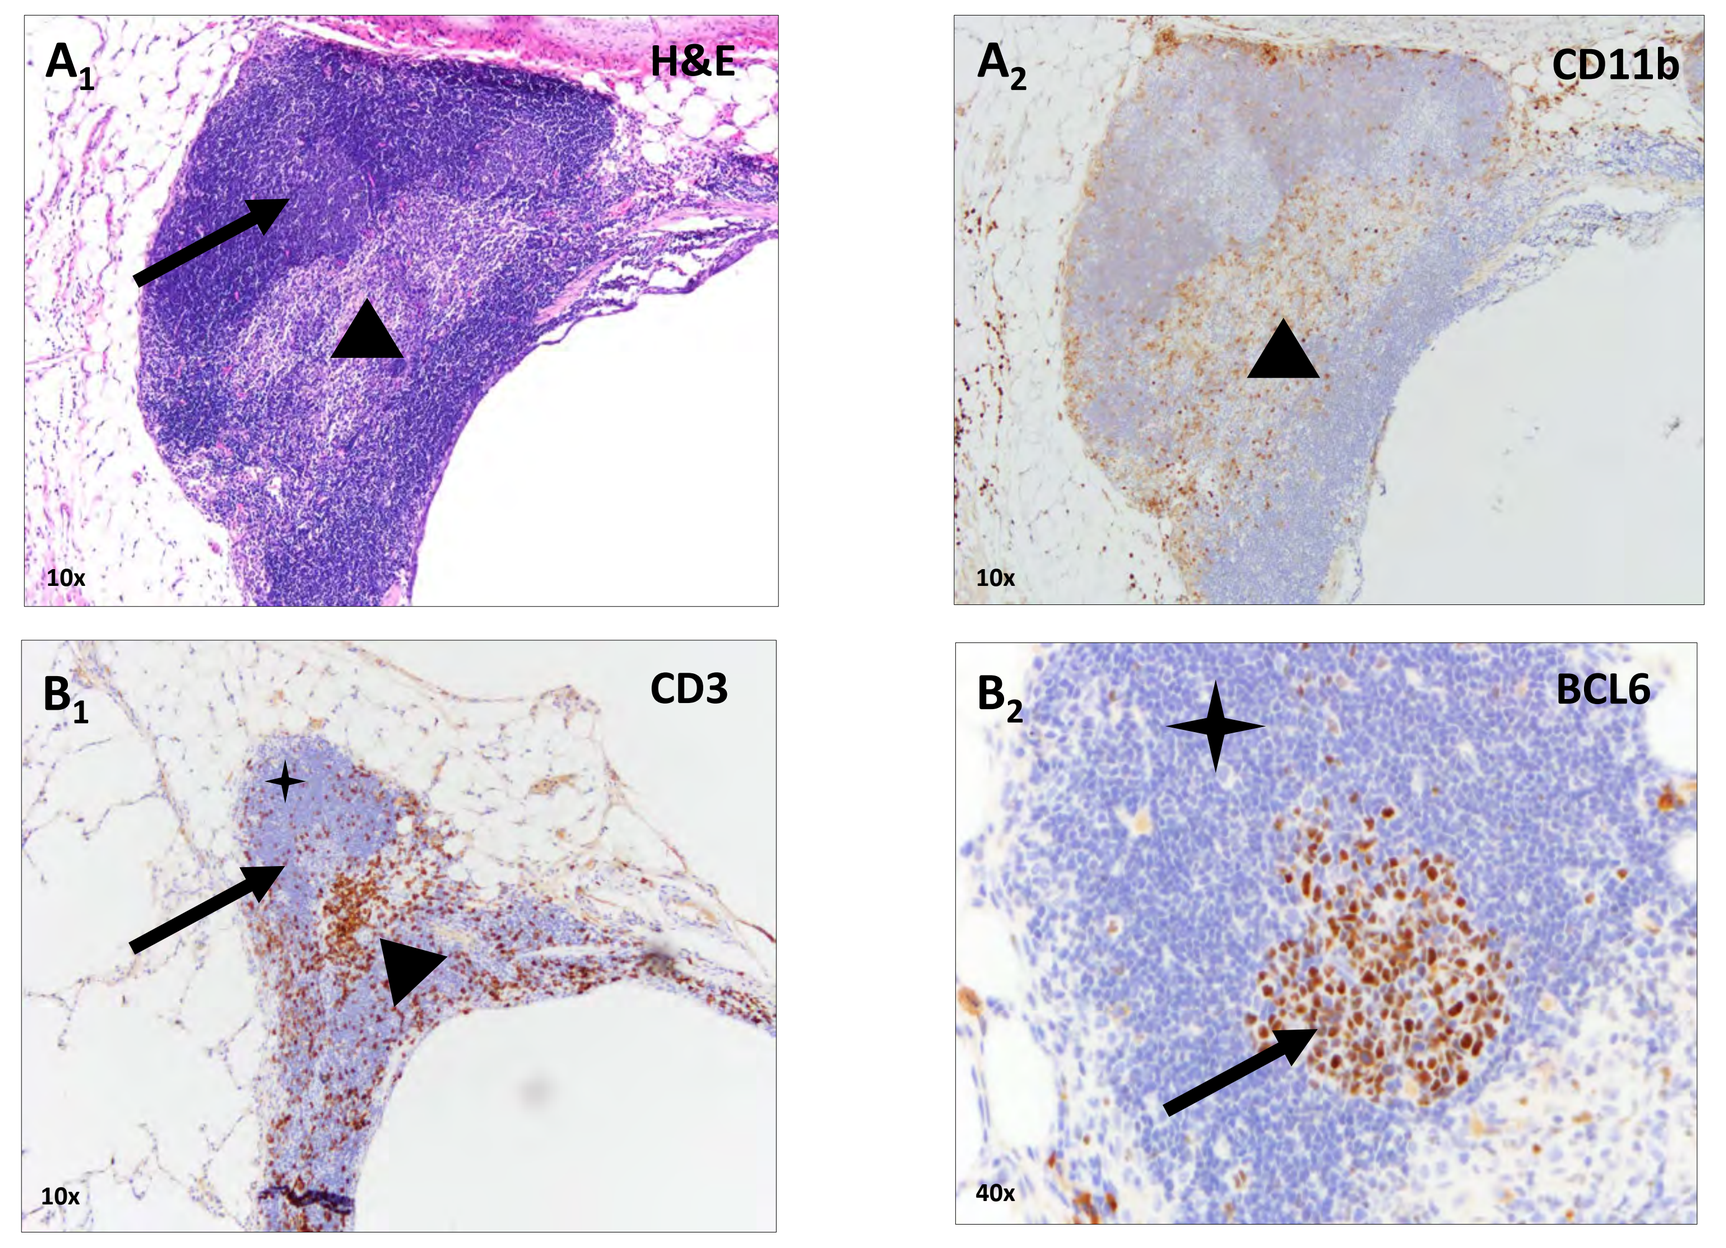
A_1 H&E 10x
A_2 CD11b 10x
B_1 CD3 10x
B_2 BCL6 40x

Top row: H&E staining (A_1) showing a tertiary lymphoid structure with an organoid appearance and lymphoid follicles composed of mantle zone B-cells (black arrow), as well as a sinus containing histocytes and lymphocytes (black triangle). CD11b staining (A_2) shows macrophages concentrated within the sinus (black triangle).

Bottom row: Tertiary lymphoid structure (B_1) with a small unstained germinal center (black arrow) and surrounding mantle zone of B-cells (star). CD3 immunostaining staining highlights sinusoidal and paracortical T-cells (black triangle). BCL-6 staining (B_2) higher power image of B_1 shows positively stained B-cells localized within the germinal center (black arrow) and unstained surrounding mantle zone B-cells (black star).

Summary and Conclusion

- Inhaled NanoPac was found to be safe in orthotopic Calu-3 lung cancer model in T-cell deficient rats.[2,3]
- Inhaled NanoPac demonstrated greater incidence of tumor regression, and in some cases saw morphological absence of residual tumor.
- H&E stained slides indicate inhaled NanoPac was associated with an immunological response of lymphocytic infiltration into lung tumors.
- CD11b⁺ immune cell infiltration and tertiary lymphoid structures were associated with tumor regression in the inhaled NanoPac groups.
- The immune stimulatory role may be in part due to prolonged, high concentration of paclitaxel in the lung[1] as analysis of the IHC subset revealed that recruitment of the endogenous immune system to infiltrate tumors was not seen to the same extent in untreated or IV nab-paclitaxel treated animals.
- Follow-on studies to identify the lymphocytic infiltration through immunohistochemical staining and flow cytometry will further characterize the kinetics of the NanoPac-induced tumoricidal immune response.
- IND enabling toxicology studies are ongoing in preparation for clinical trials.

Reduction in tumor burden and increased immune cell infiltration was also noted in studies of intratumoral administration of submicron particle docetaxel (NanoDoce®) treating genitourinary-oncologic xenografts in rats and mice.[4]

References

1. Verco J, Johnston W, Baltezor M, et al. Pharmacokinetic profile of inhaled submicron particle paclitaxel (NanoPac®) in a rodent model. J Aerosol Med Pulm Drug Deliv. 2018.
2. diZerega G, Johnston W, Verco J, Baltezor MJ, Wolff RK, Kuehl P. NanoPac inhalation treatment of NSCLC in a nude rat orthotopic lung cancer model. Journal of Clinical Oncology. 2018;36(15_suppl):8535-8535.
3. Verco J, Johnston W, Frost M, et al. Inhaled Submicron Particle Paclitaxel (NanoPac®) Induces Tumor Regression and Immune Cell Infiltration in an Orthotopic Athymic Nude Rat Model of Non-Small Cell Lung Cancer. J Aerosol Med Pulm Drug Deliv. (*provisionally accepted pending revisions*)
4. Maulhardt HA, Hylle L, Frost MV, et al. Local Injection of Submicron Particle Docetaxel is Associated with Tumor Eradication, Reduced Systemic Toxicity and an Immunologic Response in Uro-oncologic Xenografts. Cancers. 2019;11(4):577.

Contact
Gere diZerega, MD gere.dizerega@usbiotest.com
William Johnston william.johnston@usbiotest.com
NanOlogy.us